TurboCAR™ T Cells: CAR T Cells with Constitutive, Programmable Cytokine Signaling Outputs

Regina Lin[1], Janette Sutton[1], Trevor Bentley[1], Melinda Au[1], Tom Van Blarcom[1], Cesar Sommer[1], Barbra Sasu[1], Siler Panowski[1]

[1]Allogene Therapeutics, Inc., South San Francisco, CA, USA

Abstract

CAR T cell therapy has attained unprecedented success in the treatment of certain hematological malignancies in the autologous setting. However, having an allogeneic approach with a meaningful clinical benefit in hematological malignancies is desired. In addition, the clinical benefit in solid tumor indications has been limited, potentially in part due to suppressive solid tumor microenvironments (TME) that inhibit T cell effector function and persistence. While the provision of cytokine support can help CAR T cells overcome suppressive TME, combining CAR T therapy with systemically-administered cytokines/cytokine mimetics can result in toxicities and adverse events. Even cell therapies locally secreting cytokines can still activate neighboring cells and cause pleiotropic effects. To gain the benefit of cytokine stimuli without adverse effects, we designed TurboCAR™ T cells, which are CAR T cells co-expressing an inducible chimeric cytokine receptor or Constitutively Active Chimeric Cytokine Receptor (CACCR). CACCRs are homodimeric receptor chimeras that mimic cytokine signaling in a constitutive, CAR T cell-intrinsic fashion. For example, CACCRs may comprise (i) a membrane-tethered dimerization and JAK-binding domain derived from the thrombopoietin receptor (TpoR), fused to (ii) one or more intracellular signaling domains derived from the cytokine receptor(s) of interest. Mutations in the TpoR transmembrane domain result in constitutive receptor dimerization and activation of JAK2, which in turn phosphorylates the cytokine receptor signaling domains and cause STAT signaling. Each of these domains were individually optimized, and then combinatorially fused to generate CACCRs that mimicked downstream STAT signaling of cytokine receptors of interest. CACCRs mimicking signaling from multiple cytokines were generated. An EGFRvIII tool CAR was utilized to screen and select CACCRs based on TurboCAR™ T cell manufacturability and in vitro serial killing activity. Functional benefits conferred by CACCRs were further validated in the context of a CAR directed towards BCMA in a range of functional assays, demonstrating improved activity compared to the parental CAR. In conclusion, CACCRs are novel homodimeric cytokine receptor chimeras that can be tailored through fusion of dimerization and signaling domains to increase TurboCAR™ T cell activity, while circumventing safety risks associated with cytokine combination therapy.

Design of homodimeric Constitutively Active Chimeric Cytokine Receptors (CACCRs)

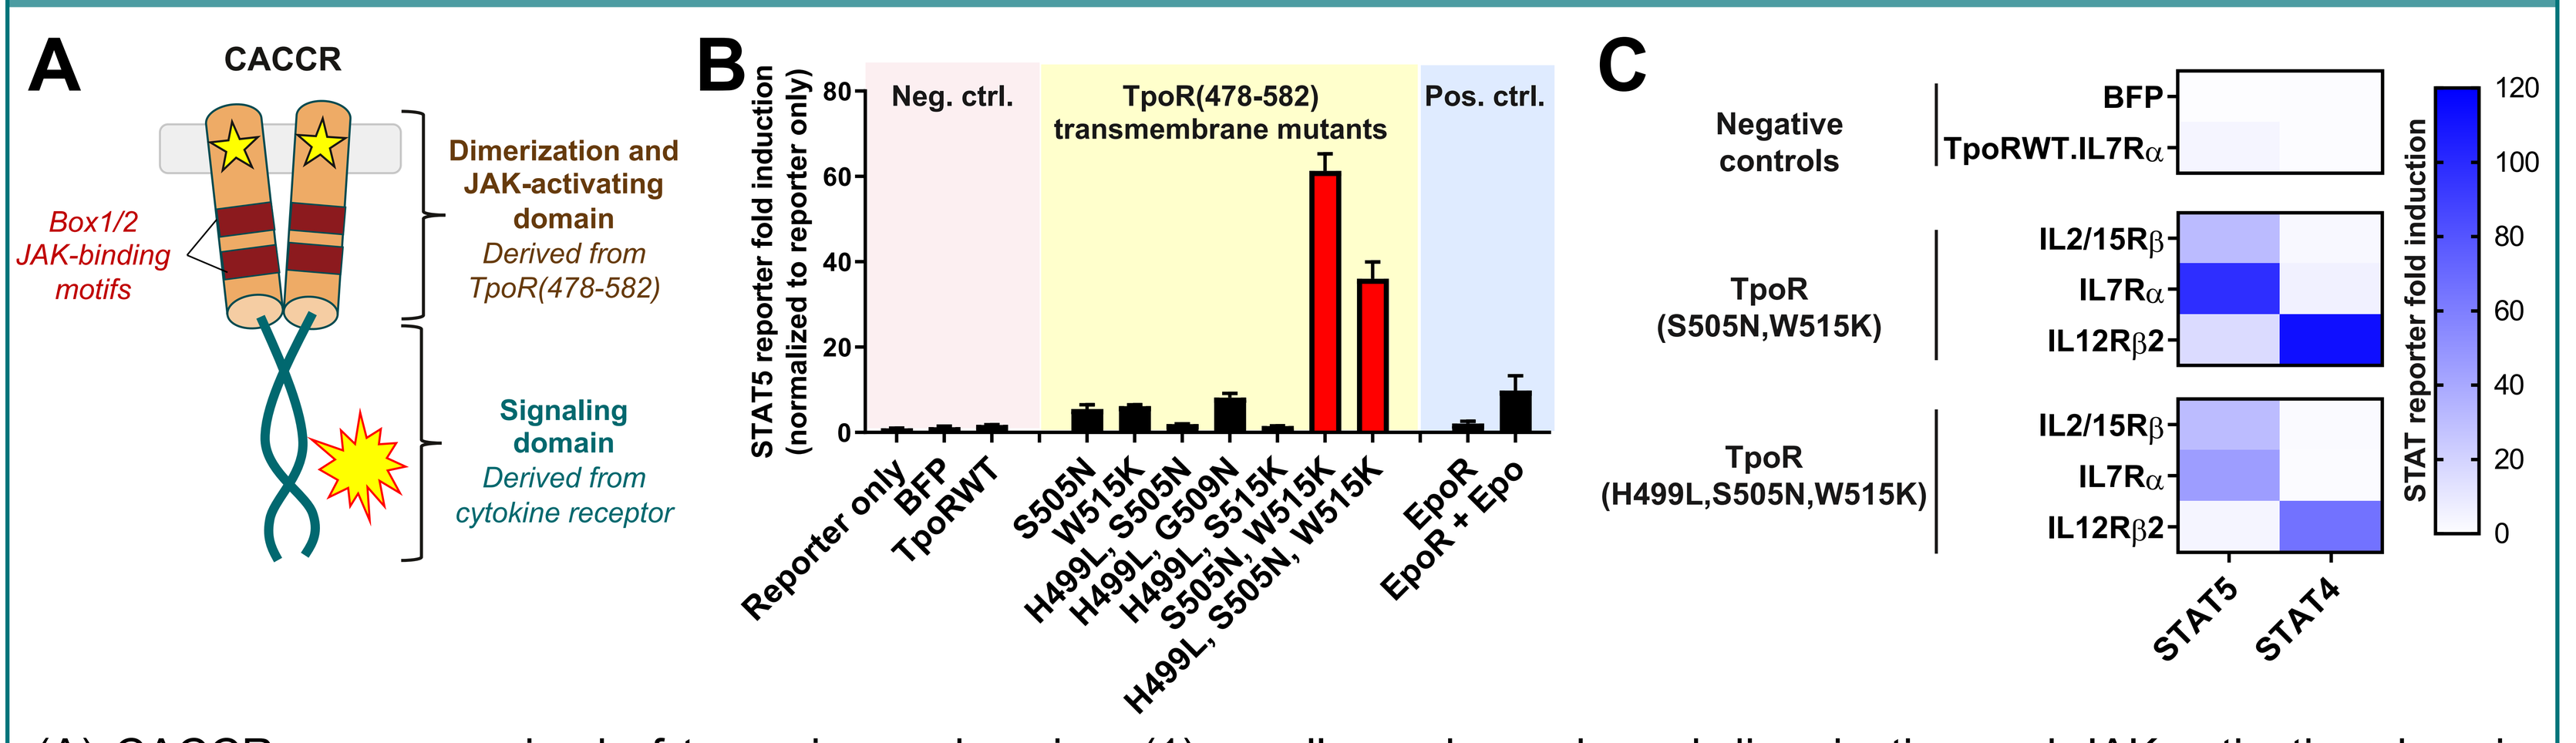
A
CACCR
Box1/2 JAK-binding motifs
Dimerization and JAK-activating domain
Derived from TpoR(478-582)
Signaling domain
Derived from cytokine receptor
B
STAT5 reporter fold induction (normalized to reporter only)
Neg. ctrl.
TpoR(478-582) transmembrane mutants
Pos. ctrl.
Reporter only
BFP
TpoRWT
S505N
W515K
H499L, S505N
H499L, G509N
H499L, S515K
S505N, W515K
H499L, S505N, W515K
EpoR
EpoR + Epo
C
Negative controls
BFP
TpoRWT.IL7Rα
TpoR (S505N,W515K)
TpoR (H499L,S505N,W515K)
IL2/15Rβ
IL7Rα
IL12Rβ2
STAT5
STAT4
STAT reporter fold induction

(A) CACCRs are comprised of two primary domains: (1) a cell membrane-bound dimerization and JAK-activating domain derived from TpoR, coupled to (2) an intracellular signaling domain containing phosphorylable tyrosine residues derived from a cytokine receptor of interest. (B) Using an IL7Rα-based CACCR, TpoR transmembrane domain mutants were screened to identify those capable of enforcing strong, constitutive receptor dimerization and signaling. (C) CACCRs bearing full-length signaling domains derived from the indicated cytokine receptors induced downstream STAT signaling expected of the parental cytokine receptor. Full-length signaling domains are IL2/15Rβ(333-551), IL7Rα(316-459) and IL12Rβ2(714-862).

CACCR signaling domains can be tailored to reduce vector cargo size and generate diverse signaling outputs

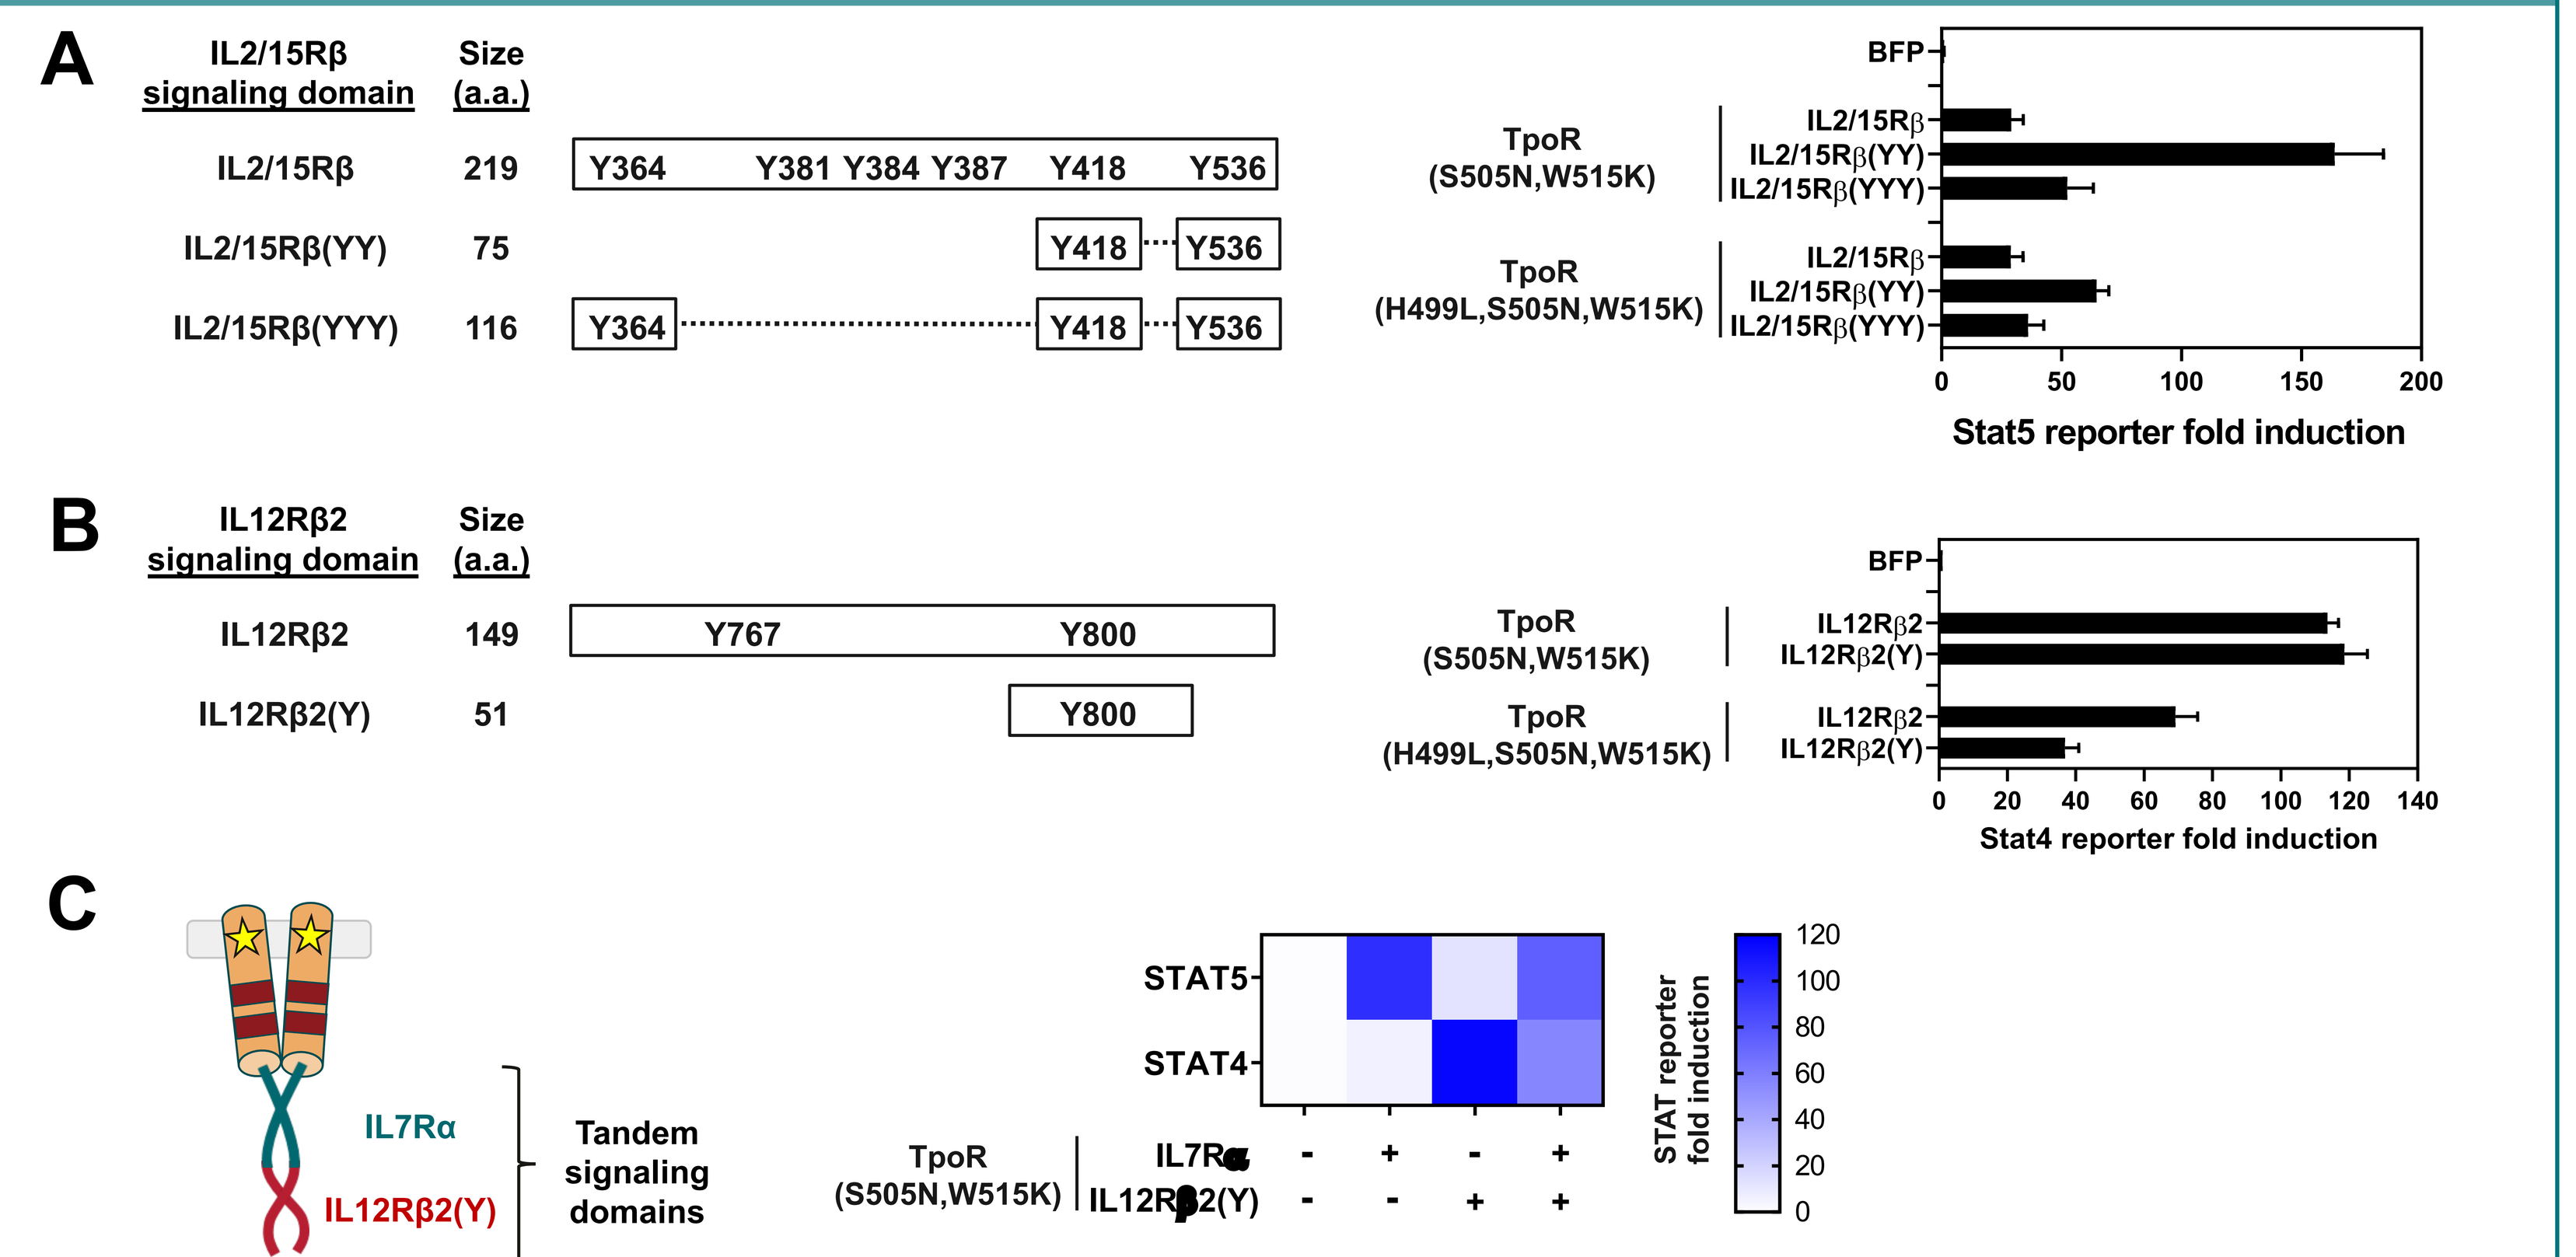
A
IL2/15Rβ signaling domain
Size (a.a.)
IL2/15Rβ 219
Y364 Y381 Y384 Y387 Y418 Y536
IL2/15Rβ(YY) 75
Y418 Y536
IL2/15Rβ(YYY) 116
Y364 Y418 Y536
TpoR (S505N,W515K)
TpoR (H499L,S505N,W515K)
Stat5 reporter fold induction
B
IL12Rβ2 signaling domain
Size (a.a.)
IL12Rβ2 149
Y767 Y800
IL12Rβ2(Y) 51
Y800
Stat4 reporter fold induction
C
IL7Rα
IL12Rβ2(Y)
Tandem signaling domains
STAT5
STAT4
TpoR (S505N,W515K)
STAT reporter fold induction

(A & B)Truncations of the full-length IL2/15Rβ(333-551) or IL12Rβ2(714-862) signaling domains were generated by retaining regions bearing only the indicated tyrosine residues. Signaling activity was assessed in a STAT reporter assay. (A) Signal strength of the full-length IL2/15Rβ(333-551) was recapitulated by the IL2/15Rβ(YYY) variant, and surpassed by the IL2/15Rβ(YY) variant. (B) The significantly smaller IL12Rβ2(Y) signaling domain activated STAT4 efficiently, with the potential to recapitulate the signal strength of full-length IL12Rβ2(714-862). (C) Signaling domains derived from distinct cytokine receptors can be fused in tandem to simultaneously activate multiple STAT pathways from a single CACCR.

IL2/15Rβ-derived CACCRs more closely mimic IL-15 signaling

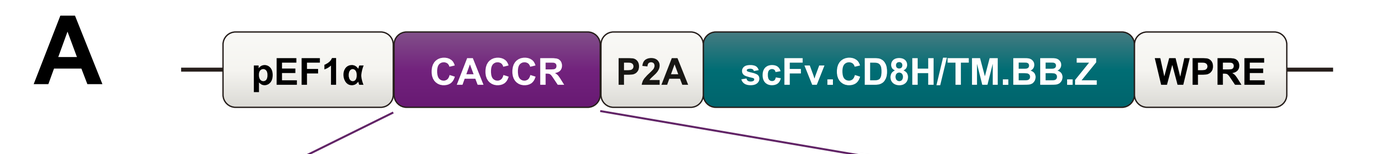
A
pEF1α
CACCR
P2A
scFv.CD8H/TM.BB.Z
WPRE

TurboCAR™	Coexpressed CACCR
TurboCAR.15.1	TpoR(S505N,W515K). IL2/15Rβ(YY)
TurboCAR.15.3	TpoR(H499L,S505N,W515K) IL2/15Rβ(YYY)

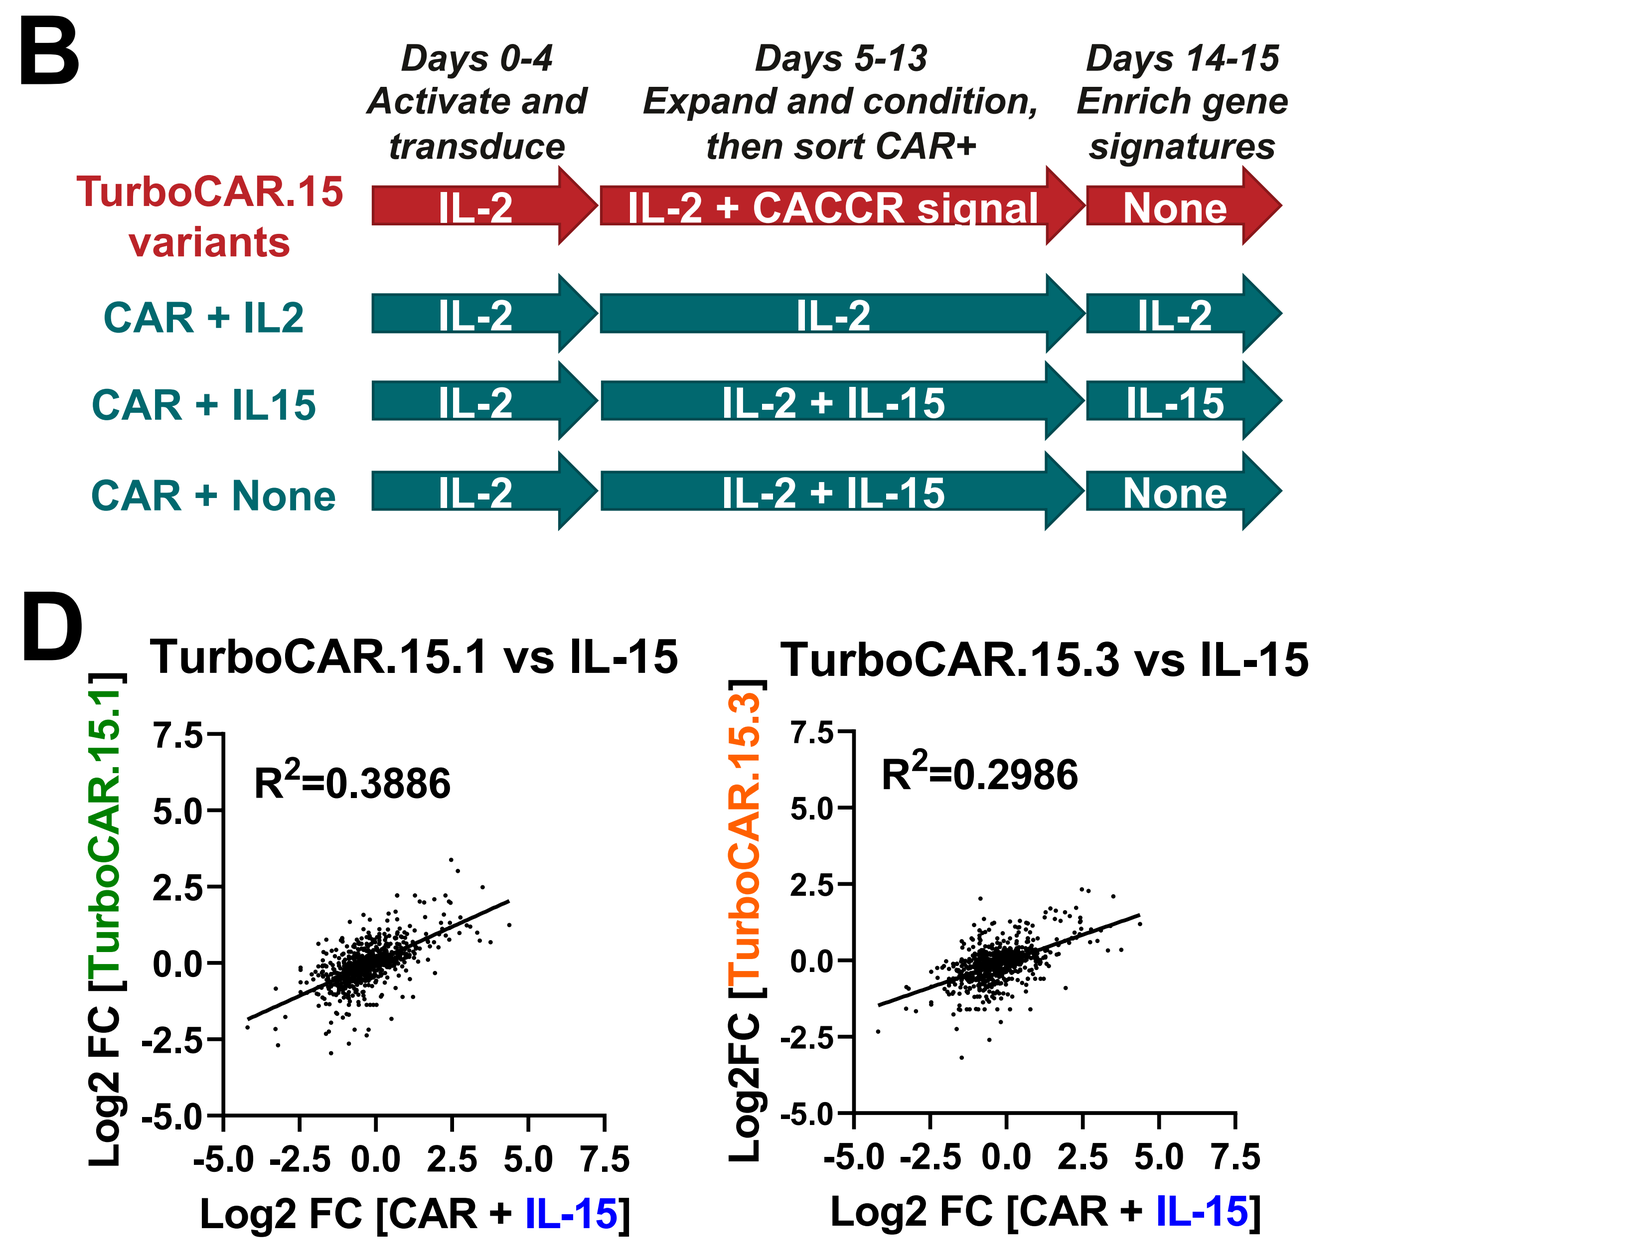
B
Days 0-4 Activate and transduce
Days 5-13 Expand and condition, then sort CAR+
Days 14-15 Enrich gene signatures
TurboCAR.15 variants
IL-2
IL-2 + CACCR signal
None
CAR + IL2
CAR + IL15
IL-2 + IL-15
IL-15
CAR + None
C
TurboCAR.15.1 vs IL-2
R^2=0.002102
TurboCAR.15.3 vs IL-2
R^2=0.0002979
Log2 FC [CAR + IL-2]
D
TurboCAR.15.1 vs IL-15
R^2=0.3886
TurboCAR.15.3 vs IL-15
R^2=0.2986
Log2 FC [CAR + IL-15]

TurboCAR.15 variants derived from the full-length IL-2/15Rβ may mimic the signaling effects of IL-2, IL-15, or a combination of both. (A) Schematic of TurboCAR construct containing the indicated IL-2/15Rβ-derived CACCRs. (B) Experimental design for gene expression analysis using the Nanostring nCounter Human CAR T panel. TurboCAR.15 T cells were generated by expansion in IL-2 as per standard protocol. To mimic the aggregate of cytokine signaling received by TurboCAR.15 T cells, unmodified CAR T cells were conditioned in either IL-2 alone, or a combination of IL-2 and IL-15. (C & D) Scatter plots showing gene expression changes in TurboCAR.15 variants compared to (C) IL-2 conditioned and (D) IL-15 conditioned CAR T cells. Log2 FC was calculated by normalization to the "CAR + None" control.

TurboCAR™ cytokine signaling is T cell-intrinsic

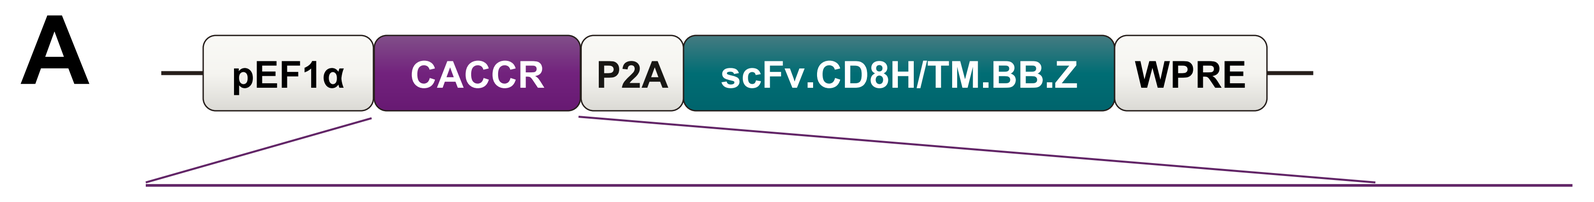
A
pEF1α
CACCR
P2A
scFv.CD8H/TM.BB.Z
WPRE

TurboCAR™	Coexpressed CACCR	% CAR+ & yield*
TurboCAR.7.FL	TpoR(H499L,S505N,W515K). IL7Rα(316-459)	≥ unmodified CAR
TurboCAR.15.FL	TpoR(S505N,W515K). IL2/15Rβ(333-551)	< unmodified CAR
TurboCAR.15.1	TpoR(S505N,W515K). IL2/15Rβ(YY)	≥ unmodified CAR
TurboCAR.15.2	TpoR(H499L,S505N,W515K) IL2/15Rβ(YY)	≥ unmodified CAR
TurboCAR.15.3	TpoR(H499L,S505N,W515K) IL2/15Rβ(YYY)	≥ unmodified CAR
TurboCAR.12.FL	TpoR(S505N,W515K) IL12Rβ2(714-862)	< unmodified CAR
TurboCAR.12.1	TpoR(S505N,W515K) IL12Rβ2(Y)	< unmodified CAR

* ≥ unmodified CAR; < unmodified CAR

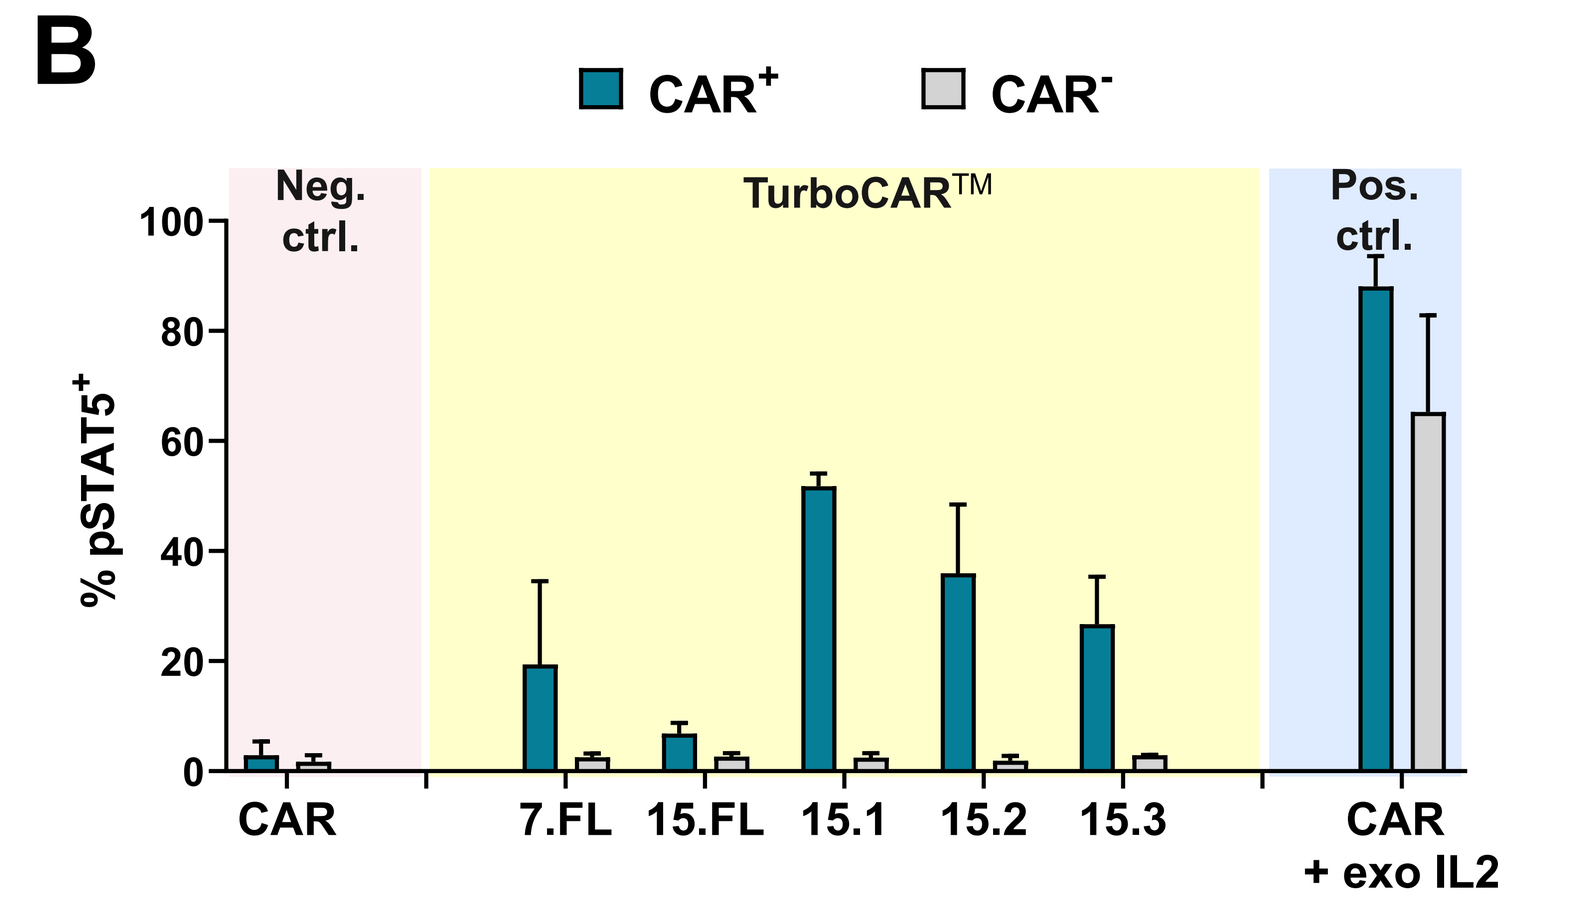
B
CAR+
CAR-
% pSTAT5+
Neg. ctrl.
TurboCAR™
Pos. ctrl.
CAR
7.FL
15.FL
15.1
15.2
15.3
CAR + exo IL2

(A) Schematic of TurboCAR construct and list of selected TurboCAR abbreviations coexpressing the indicated CACCR formats. Manufacturability of each TurboCAR relative to the unmodified CAR is shown. (B) CACCR signaling activity was assessed in human TurboCAR T cells bearing the EGFRvIII-specific 2173 scFv by intracellular flow cytometry staining for pSTAT5. STAT5 activation was detected only in the CAR⁺, but not CAR⁻ fraction of TurboCAR-transduced T cells, demonstrating TurboCAR T cell-intrinsic cytokine signaling. By contrast, CAR T cells that had been treated with exogenous IL2 showed STAT5 activation in both CAR⁺ and CAR⁻ fractions.

TurboCAR™ signaling domains enhance CAR T cell cytotoxicity

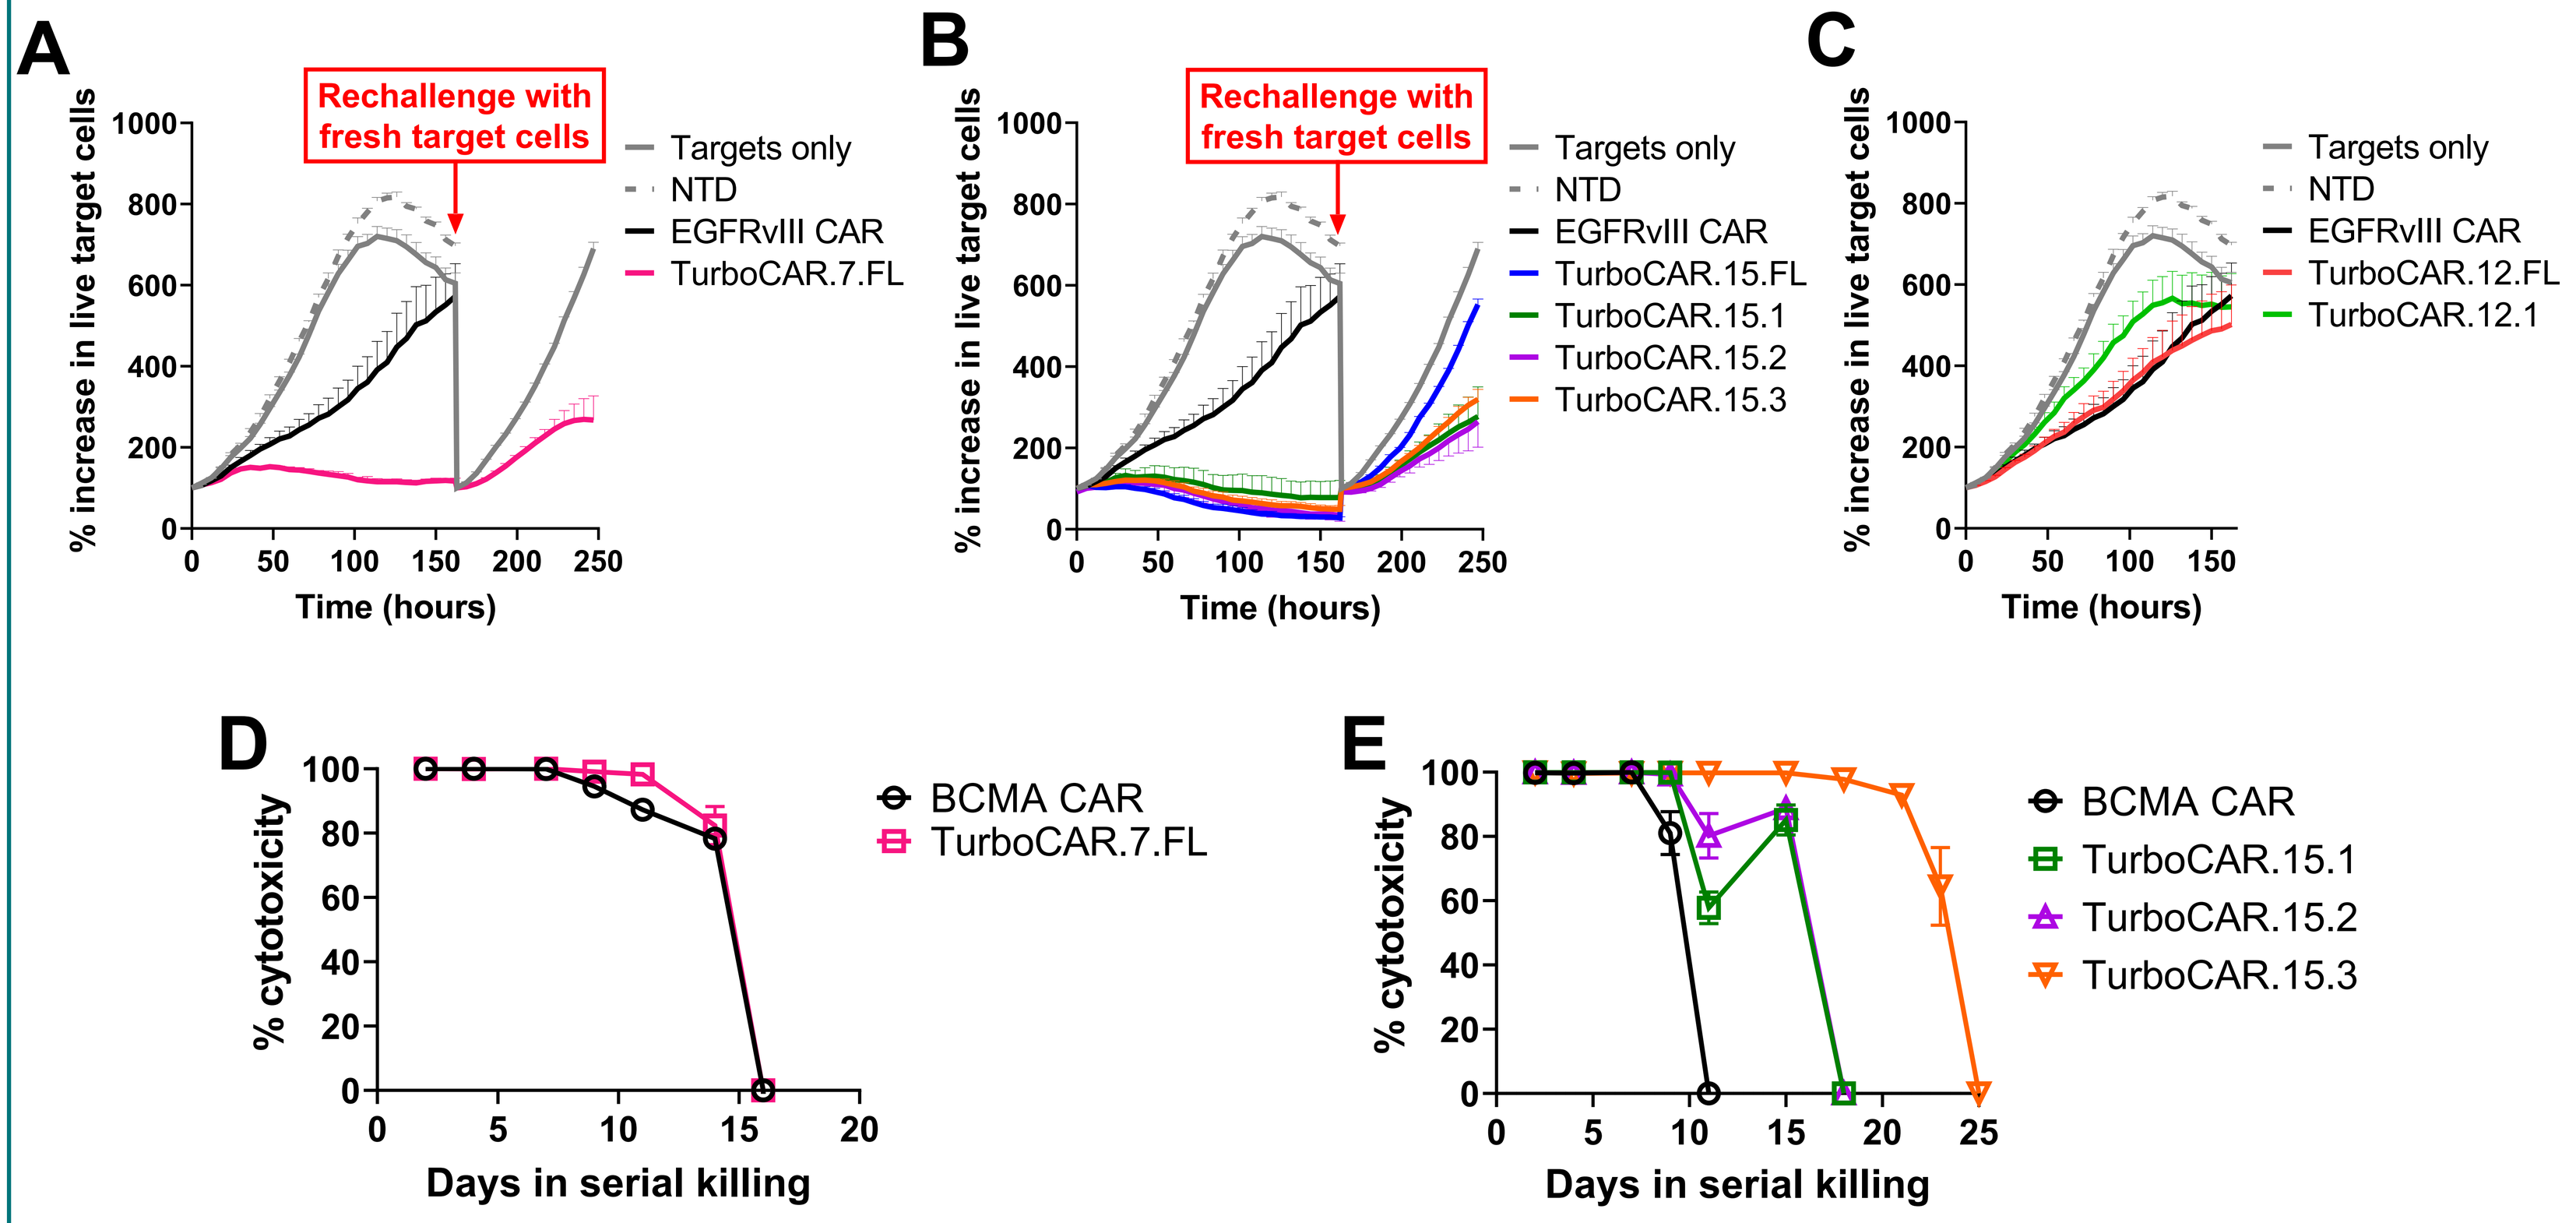
A
Rechallenge with fresh target cells
Targets only
NTD
EGFRvIII CAR
TurboCAR.7.FL
% increase in live target cells
Time (hours)
B
TurboCAR.15.FL
TurboCAR.15.1
TurboCAR.15.2
TurboCAR.15.3
C
TurboCAR.12.FL
TurboCAR.12.1
D
BCMA CAR
TurboCAR.7.FL
% cytotoxicity
Days in serial killing
E
BCMA CAR
TurboCAR.15.1
TurboCAR.15.2
TurboCAR.15.3

(A-C) Cytotoxicity of TurboCARs bearing the EGFRvIII-specific 2173 scFv and the indicated CACCRs were evaluated using U87-EGFRvIII-nucGFP target cells at an E:T of 1:3. Where indicated, EGFRvIII TurboCAR T cells were rechallenged with fresh target cells. Live target counts were monitored using the Incucyte Live Cell Imaging System. (A) TurboCAR.7.FL and (B) TurboCAR.15 variants showed enhanced initial potency, as well as long-term durability of cytotoxic responses. (C) TurboCAR.12 variants did not enhance the activity of CAR T cells in vitro, and were therefore excluded from further testing. (D & E) Long-term cytotoxicity of TurboCARs directed towards BCMA was assessed using MM1.S-Luc-GFP target cells at an E:T of 10:1. Fresh target cells were added every 2-3 days and target cell killing was determined by luminescence readout. (D) While TurboCAR.7.FL did not enhance activity, (E) TurboCAR.15 variants showed enhanced long-term cytotoxicity.

TurboCAR™ T cells show superior expansion, survival and improved functional output following repeated target stimulation

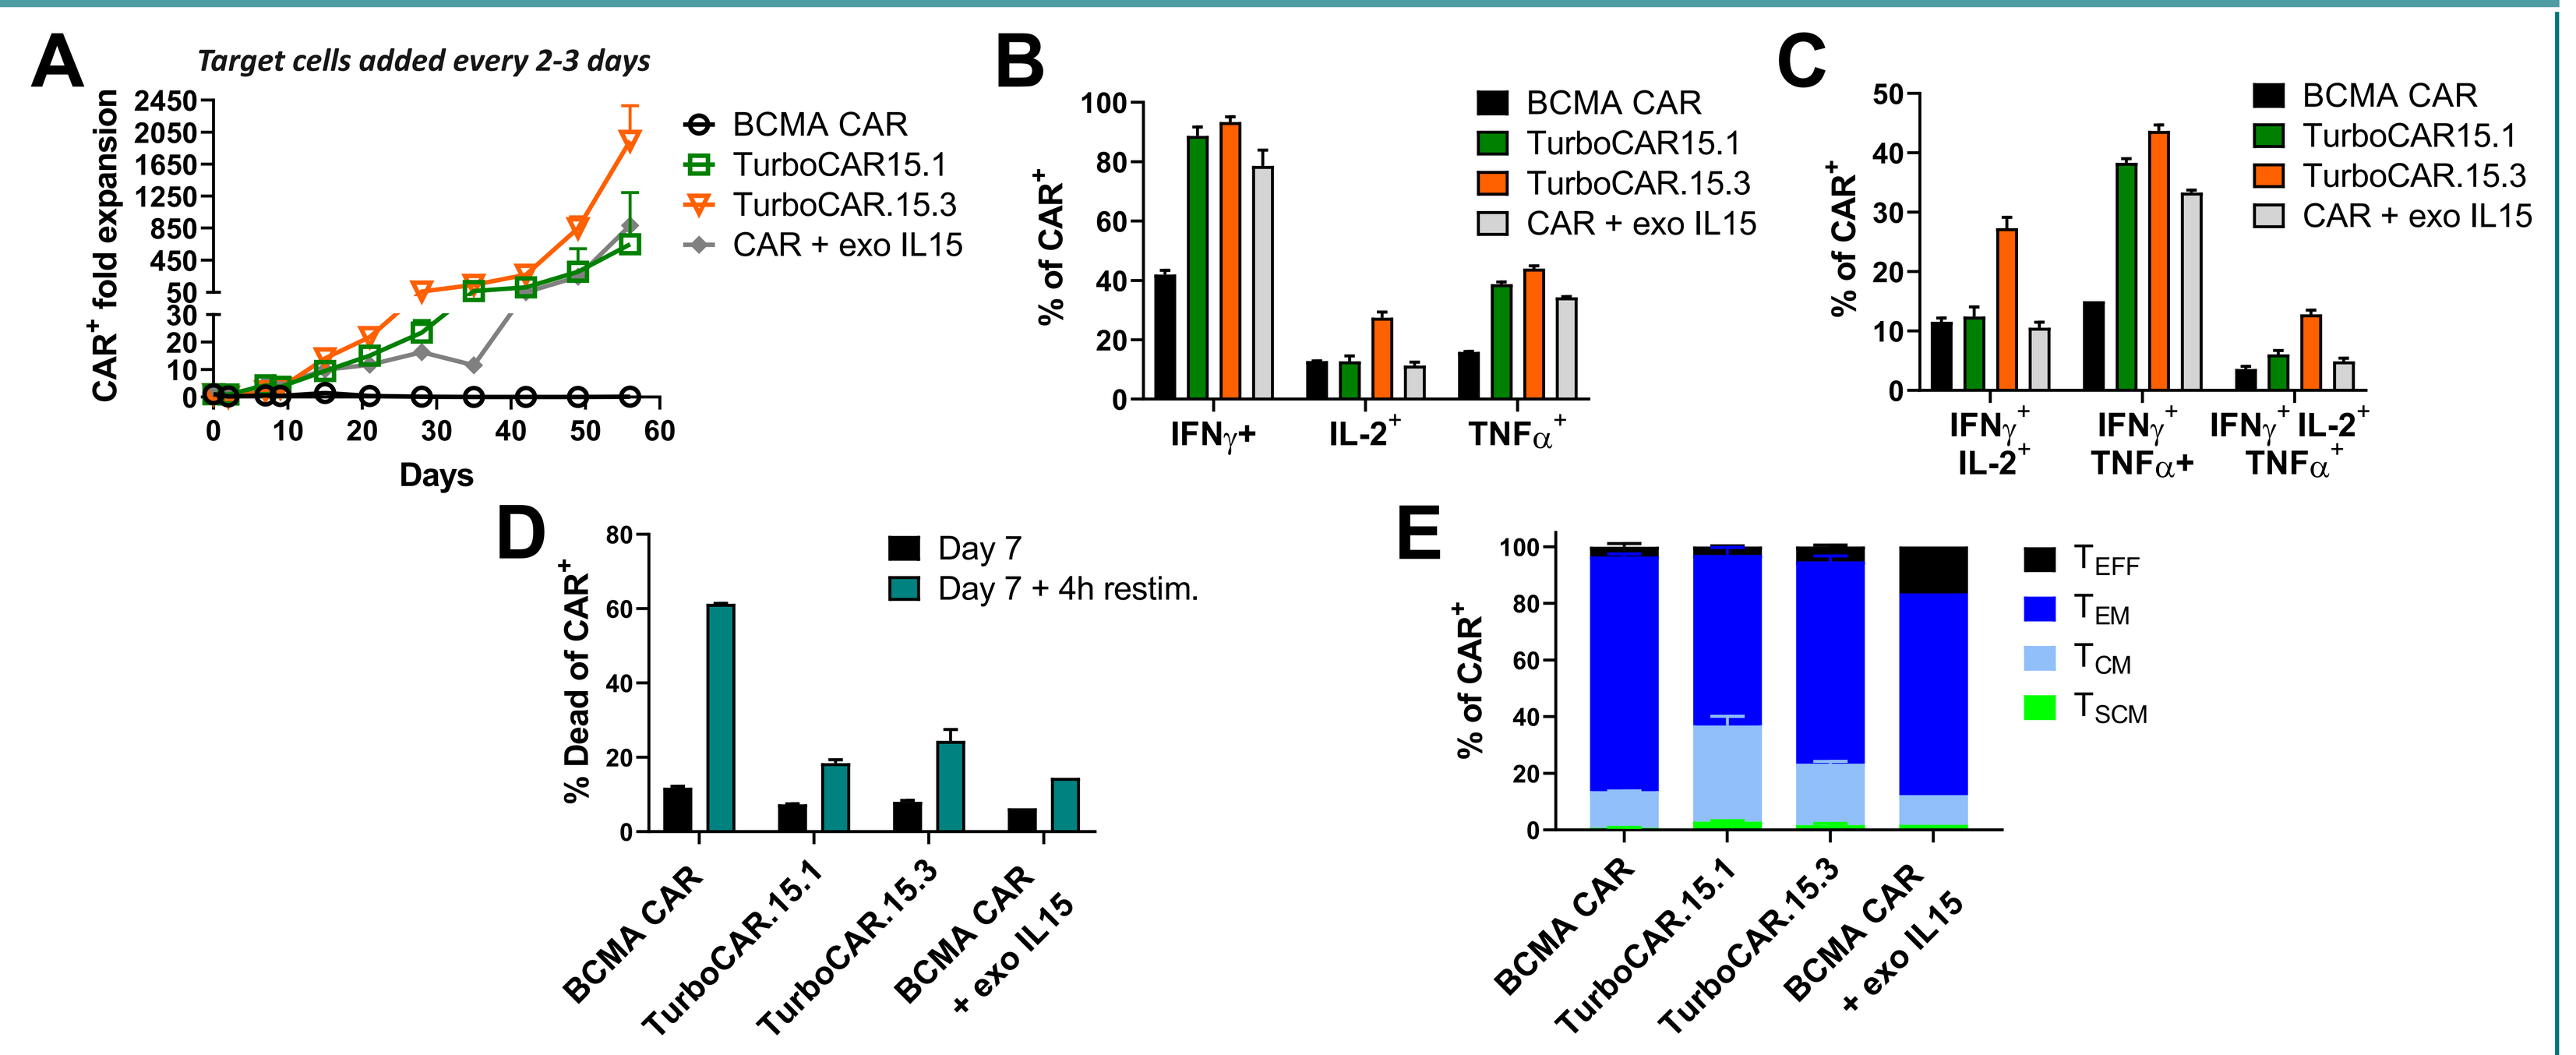
A
Target cells added every 2-3 days
CAR+ fold expansion
Days
BCMA CAR
TurboCAR15.1
TurboCAR.15.3
CAR + exo IL15
B
% of CAR+
IFNγ+
IL-2+
TNFα+
C
D
% Dead of CAR+
Day 7
Day 7 + 4h restim.
E
T_{EFF}
T_{EM}
T_{CM}
T_{SCM}

(A) On-target expansion of TRAC/CD52 dKO BCMA CAR T cells serially challenged with MM1.S-Luc-GFP targets. (B-D) On Day 7, CAR T cells were evaluated for intracellular cytokine expression and activation-induced cell death (AICD). TurboCAR T cells showed (B) improved expression of effector cytokines, (C) greater polyfunctionality and (D) were protected from AICD. (D) Analysis on Day 14 revealed an improved memory phenotype and delayed differentiation of TurboCAR T cells.

Activity and survival of TurboCAR™ T cells is target-dependent

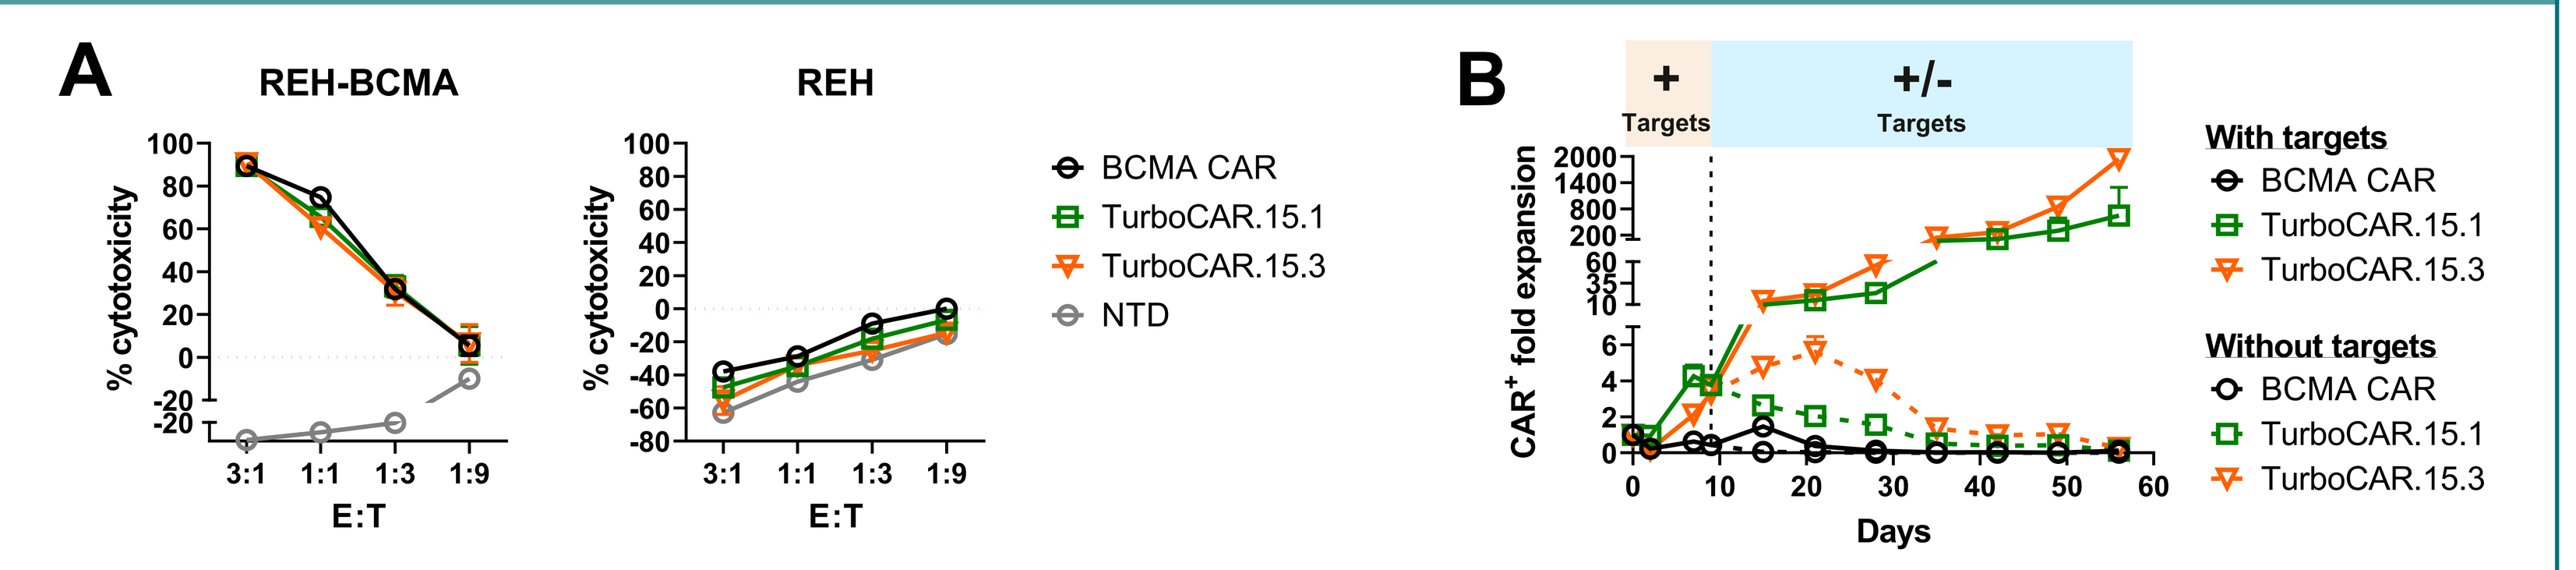
A
REH-BCMA
REH
% cytotoxicity
E:T
BCMA CAR
TurboCAR.15.1
TurboCAR.15.3
NTD
B
+ Targets
+/- Targets
CAR+ fold expansion
Days
With targets
Without targets

(A) TRAC/CD52 dKO BCMA TurboCAR T cells lyse BCMA-overexpressing REH target cells, but are inactive against the BCMA-negative parental line. (B) TRAC/CD52 dKO BCMA TurboCAR T cells were repeatedly exposed to MM1.S target cells every 2-3 days for the first 9 days to induce expansion. Following that, MM1.S target cells were either continually added or withdrawn from these cultures. Without targets, TurboCAR T cells ceased expansion and underwent a gradual decline.

TurboCAR™ T cells show enhanced anti-tumor efficacy and persistence

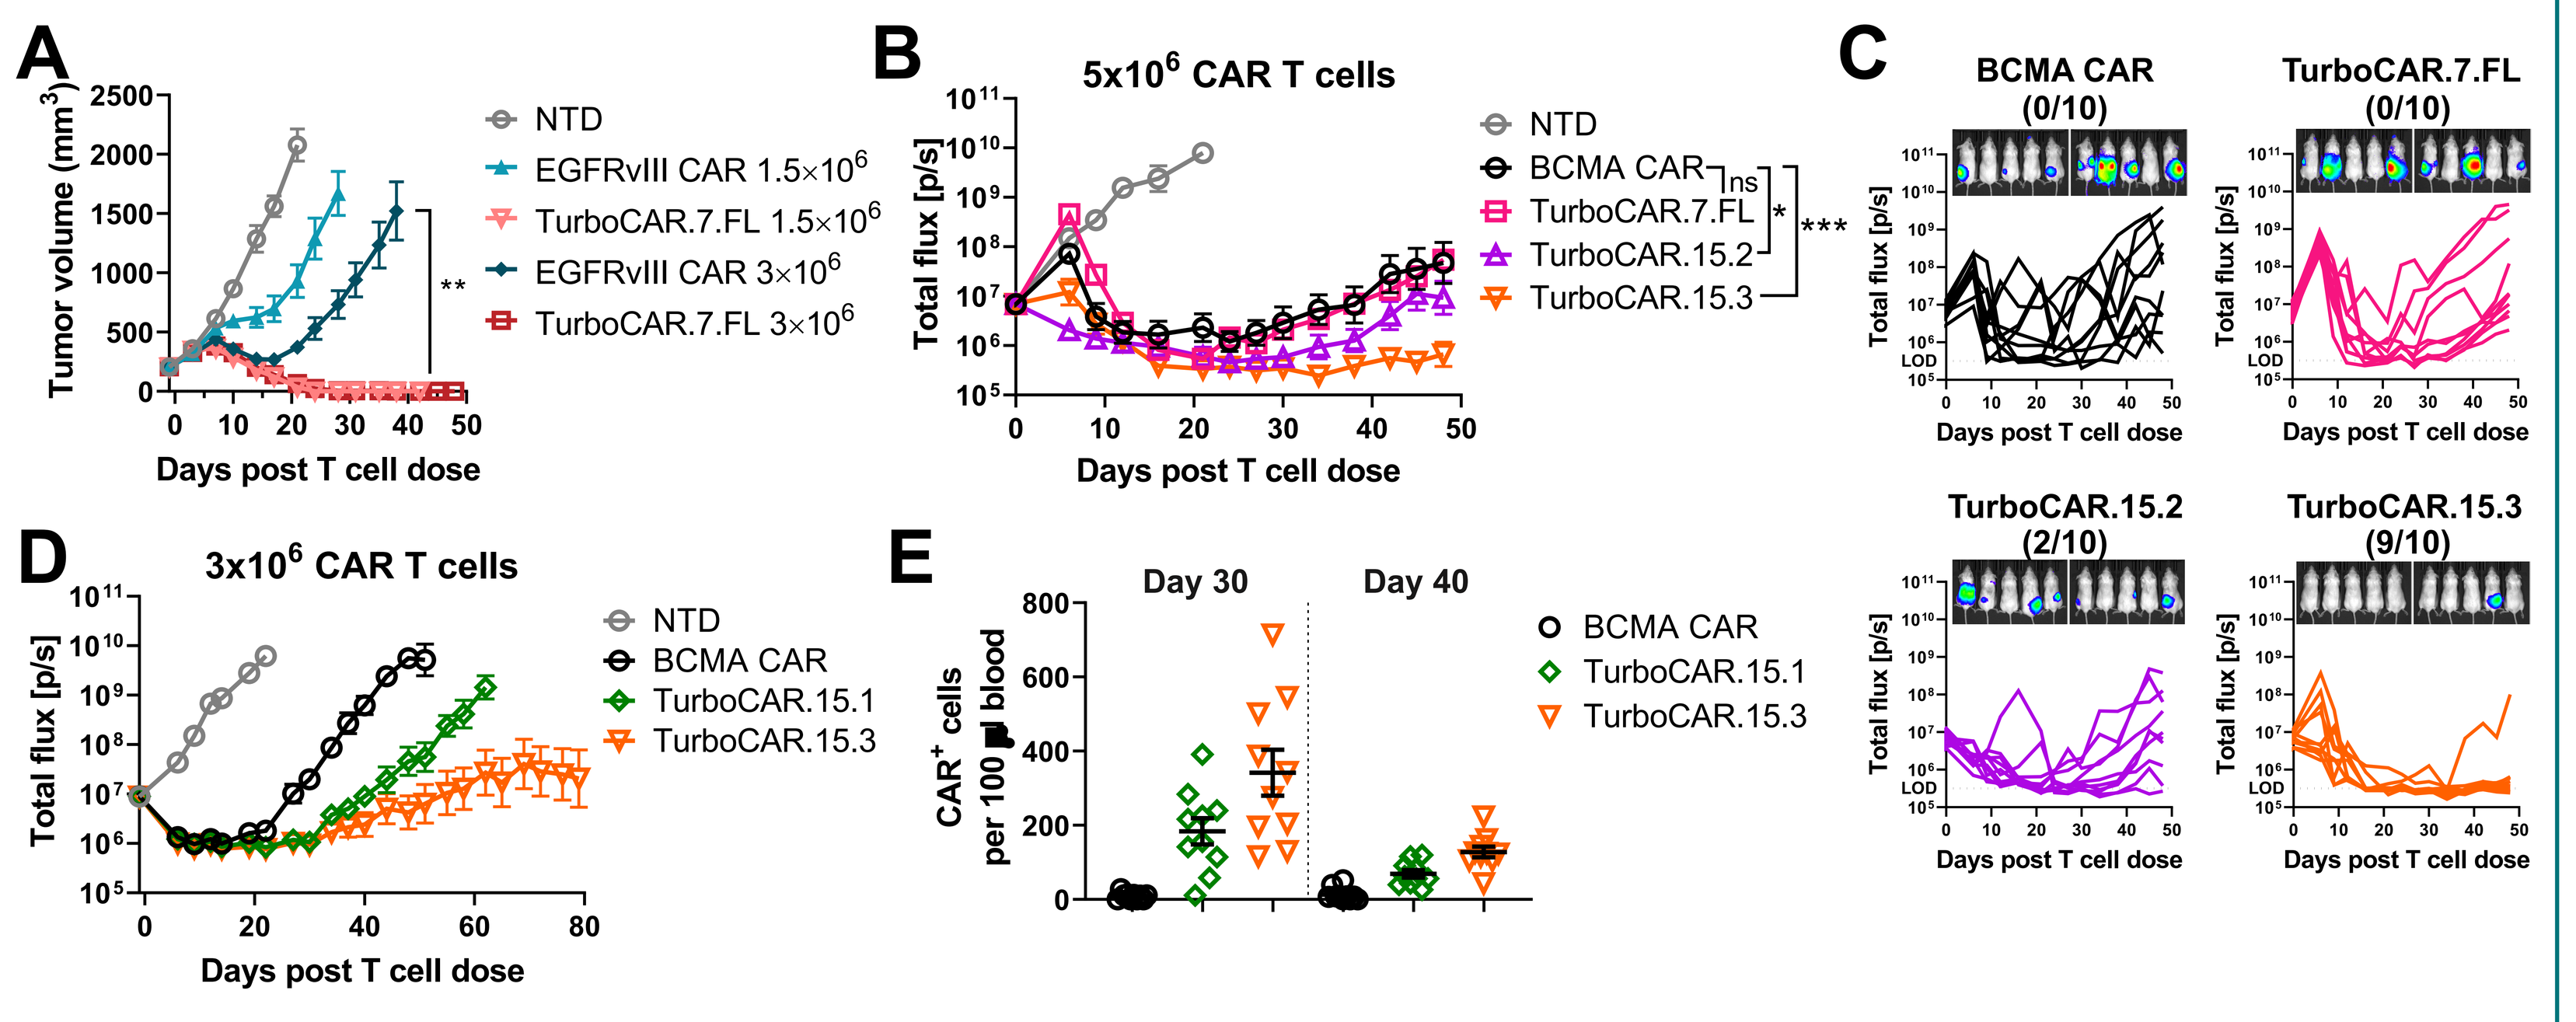
A
Tumor volume (mm³)
Days post T cell dose
NTD
EGFRvIII CAR 1.5×10⁶
TurboCAR.7.FL 1.5×10⁶
EGFRvIII CAR 3×10⁶
TurboCAR.7.FL 3×10⁶
B
5x10⁶ CAR T cells
Total flux [p/s]
NTD
BCMA CAR
TurboCAR.7.FL
TurboCAR.15.2
TurboCAR.15.3
C
BCMA CAR (0/10)
TurboCAR.7.FL (0/10)
TurboCAR.15.2 (2/10)
TurboCAR.15.3 (9/10)
D
3x10⁶ CAR T cells
TurboCAR.15.1
E
Day 30
Day 40
CAR+ cells per 100 µl blood

(A) In vivo anti-tumor activity of EGFRvIII TurboCAR T cells was assessed in NSG mice bearing established subcutaneous LN229-EGFRvIII tumors, and intravenously infused with the indicated doses of CAR T cells. While unmodified CAR T cells delayed tumor progression, TurboCAR T cells completely eliminated tumors. (B-D) Efficacy of TRAC/CD52 dKO BCMA TurboCAR T cells was assessed in an orthotopic model of multiple myeloma using the MM1.S-Luc-GFP cell line. (B) At a dose of 5×10⁶ CAR T cells, TurboCAR.15 variants prolonged the durability of anti-tumor responses, with (C) TurboCAR.15.3 preventing extramedullary relapse in 9/10 mice. Numbers in parenthesis represent the number of tumor-free mice. (D) A lower dose of 3×10⁶ CAR T cells better differentiated the efficacy of TurboCAR.15 variants. (E) On the indicated days, CAR T cells in the peripheral blood of mice from (D) were enumerated. Increased persistence of TurboCAR.15 variants correlated with improved anti-tumor efficacy.

Conclusions

- TurboCARs coexpress a homodimeric CACCR that confers constitutive, T cell-intrinsic cytokine signaling
- TurboCARs can be tailored for diverse, programmable and combinatorial signaling outputs
- TurboCAR T cells provide increased functional potency and persistence following chronic target exposure
- Despite improved expansion and persistence, TurboCAR T cell activity and survival is target-dependent
- In hematologic and solid tumor models, TurboCAR T cells enhanced anti-tumor activity in vivo

Allogene's AlloCAR T programs utilize Cellectis technology. The EGFRvIII AlloCAR T program is licensed exclusively from Cellectis by Allogene. Allogene holds exclusive U.S. rights and has granted to Servier rights to EGFRvIII product candidates for all other countries. The BCMA AlloCAR T program is licensed exclusively from Cellectis and Allogene holds global development and commercial rights to this AlloCAR T program.

RESEARCH POSTER PRESENTATION DESIGN © 2019
www.PosterPresentations.com