ALPHA3: A Pivotal Phase 2 Study of First-Line (1L) Consolidation With Cemacabtagene Ansegedleucel (Cema-Cel) in Patients (Pts) With Large B-Cell Lymphoma (LBCL) and Minimal Residual Disease (MRD) After Response to Standard Therapy

Poster TPS7085

Jason Westin, MD[1]; Gary Simmons, DO, MSHA[2]; Nancy L. Bartlett, MD[3]; Houston Holmes, MD, MBA[4]; Matthew Matasar, MD[5]; Habte Yimer, MD[6]; Mitul Gandhi, MD[7]; Jeff Sharman, MD[8]; Ran Reshef, MD, MSc[9]; Akil Merchant, MD[10]; Yuliya Linhares, MD[11]; Don Stevens, MD[12]; Alex F. Herrera, MD[13]; Frederick L. Locke, MD[14]; Amy Feng, PhD[15]; Lynn Navale, MS[15]; Jennifer Tsai, MD[15]; John B. Le Gall, MD, MBA[15]; John M. Burke, MD[16] on behalf of the ALPHA3 investigators

[1]University of Texas MD Anderson Cancer Center, Houston, TX, USA; [2]Virginia Oncology Associates, Norfolk, VA, USA; [3]Siteman Cancer Center, Washington University School of Medicine, St. Louis, MO, USA; [4]Texas Oncology/Baylor Sammons Cancer Center, Dallas, TX, USA; [5]Rutgers Cancer Institute, New Brunswick, NJ, USA; [6]Texas Oncology, Tyler, TX, USA; [7]Virginia Cancer Specialists, Gainesville, VA, USA; [8]Willamette Valley Cancer Institute, Eugene, OR, USA; [9]Columbia University Irving Medical Center, New York, NY, USA; [10]Cedars Sinai Medical Center, Los Angeles, CA, USA; [11]Baptist Health, Miami Cancer Institute, Miami, FL, USA; [12]Norton Cancer Institute, Louisville, KY, USA; [13]City of Hope National Medical Center, Duarte, CA, USA; [14]Moffit Cancer Center, Tampa, FL, USA; [15]Allogene Therapeutics, San Franscisco, CA, USA; [16]US Oncology Hematology Research Program, Rocky Mountain Cancer Centers, Aurora, CO, USA

INTRODUCTION

- Approximately 60% of patients with LBCL who receive standard 1L therapy achieve cure; however, ~30% of patients in remission at the end of therapy are expected to relapse within 2 years[1-3]
- Through PhasED-Seq–powered MRD testing (**Figure 1**), patients at highest risk of relapse can be identified, which will enable changing the current lymphoma treatment paradigm from watching and waiting for relapse to consolidating therapy for cure
- Cemacabtagene ansegedleucel (cema-cel) is an immediately available, off-the-shelf, allogeneic CD19 CAR T cell product (**Figure 2**) that has shown potent anti-tumor activity with manageable safety in phase 1 trials of patients with relapsed/refractory LBCL[4,5] and is a promising agent for consolidation in this time-sensitive treatment setting
- Treating patients with LBCL and low disease burden is favorable because it ensures treatment prior to aggressive relapse or development of new comorbidities that could preclude treatment in later lines. Additionally, this treatment setting has been shown to lead to more favorable efficacy outcomes and fewer treatment-related toxicities[6-8]
- Here, we describe the design of the pivotal ALPHA3 (NCT06500273) phase 2 study, the first randomized, open-label study to assess a CAR T cell therapy as a consolidation strategy in patients with detectable MRD measured by circulating tumor DNA (ctDNA)-based testing after standard 1L immunochemotherapy

Figure 1: PhasED-Seq Technology

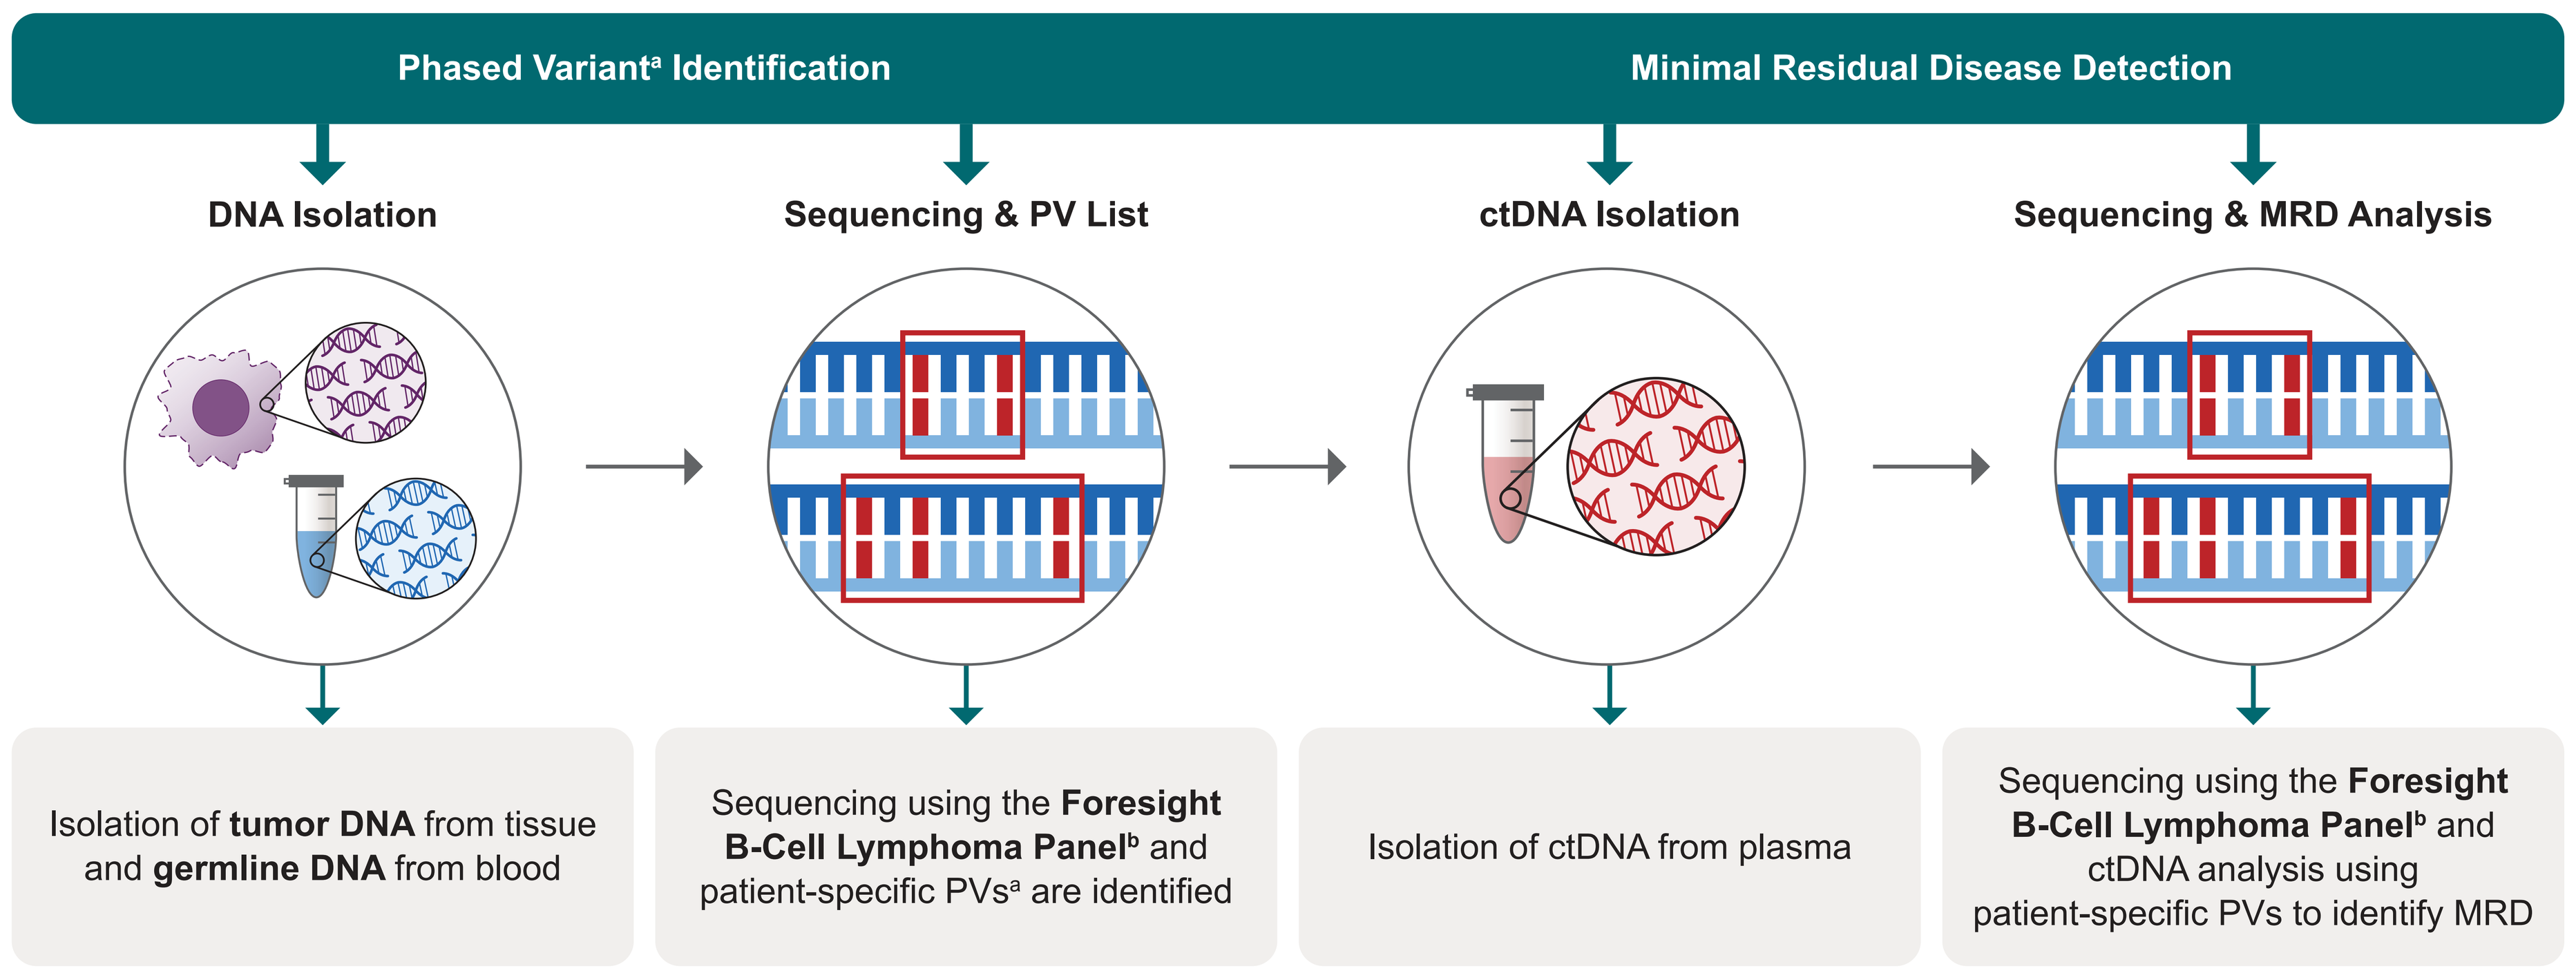
Phased Variant[a] Identification
Minimal Residual Disease Detection
DNA Isolation
Sequencing & PV List
ctDNA Isolation
Sequencing & MRD Analysis
Isolation of **tumor DNA** from tissue and **germline DNA** from blood
Sequencing using the **Foresight B-Cell Lymphoma Panel**[b] and patient-specific PVs[a] are identified
Isolation of ctDNA from plasma
Sequencing using the **Foresight B-Cell Lymphoma Panel**[b] and ctDNA analysis using patient-specific PVs to identify MRD

[a] PVs are 2 or more single nucleotide variants identified in close proximity on the same DNA molecule. [b] The panel leverages established mutational hot-spots for B-cell malignancies.
ctDNA, circulating tumor DNA; MRD, minimal residual disease; PV, phased variant.

Figure 2: Cemacabtagene Ansegedleucel (Cema-Cel)[a]

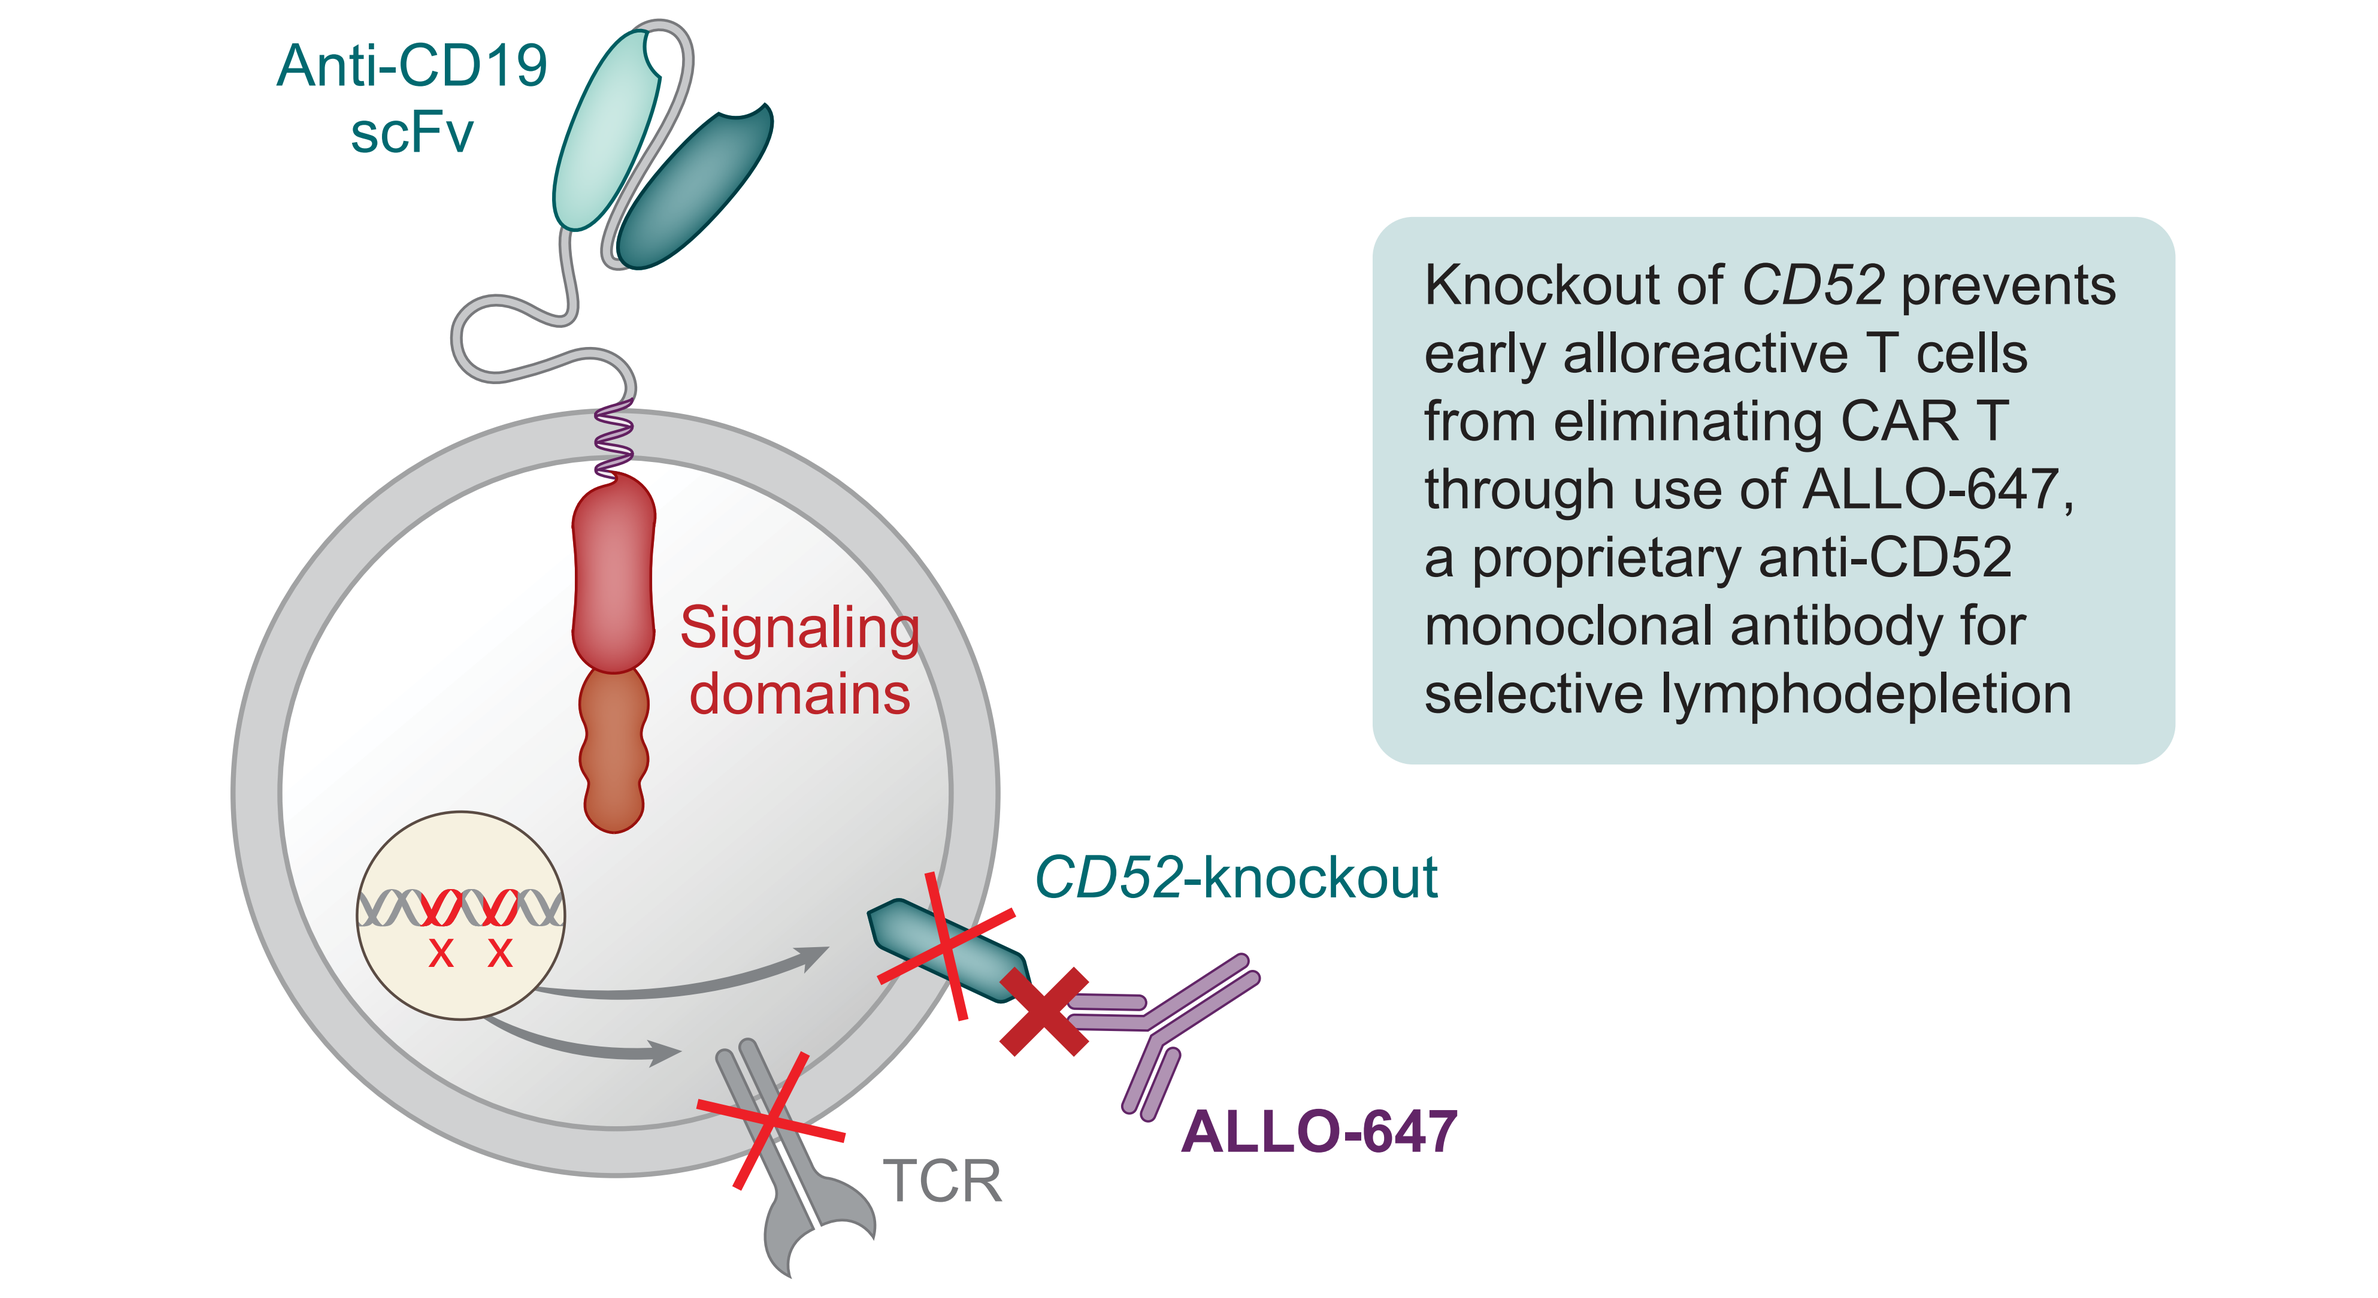
Anti-CD19 scFv
Signaling domains
CD52-knockout
ALLO-647
TCR
Knockout of CD52 prevents early alloreactive T cells from eliminating CAR T through use of ALLO-647, a proprietary anti-CD52 monoclonal antibody for selective lymphodepletion

[a] Utilizes Cellectis technologies.
CAR, chimeric antigen receptor; scFv, single chain fragment variable; TCR, T-cell receptor.

METHODS

- The ALPHA3 study will consist of a 2-part seamless design (**Figure 3**):
 - Part A
 - Randomized patients will be followed in the standard-of-care observation arm or in either one of the treatment arms (cema-cel [120×10^6 CAR T cells] following 3-day standard fludarabine [30 mg/m^2/day] and cyclophosphamide [300 mg/m^2/day] lymphodepletion with or without the anti-CD52 monoclonal antibody, ALLO-647 [30 mg/day]) in a 1:1:1 ratio
 - The selected lymphodepletion regimen for Part B will be chosen after an early interim analysis, at which time emerging data will be assessed
 - Part B
 - Randomized patients will receive treatment with the selected lymphodepletion regimen or be followed in the observation arm until the completion of accrual in a 1:1 ratio
 - An interim efficacy analysis and a primary analysis will be performed
- Treatment can occur in the inpatient or outpatient setting
- Study endpoints are listed in **Table 1**
- To be eligible to screen for the ALPHA3 study, patients must successfully complete prescreening (**Figure 4**, **Table 2**):
 - Posttreatment positron emission tomography/computed tomography (PET/CT) must show complete response or partial response for which the standard of care would be close observation (eg, negative biopsy of PET-avid lesion)
 - ctDNA-based MRD testing requires a tumor sample from initial diagnosis and a blood sample collected at or shortly after end-of-treatment PET/CT
 - Only MRD+ patients may proceed to screening and undergo further eligibility assessments

Table 1: Study Endpoints

Primary
• EFS per IRC[a]
Secondary
• PFS per IRC[a,b]
• OS[b]
• Rate of MRD clearance
• EFS per investigator[a]
• PFS per investigator[a]
• Safety (cema-cel and ALLO-647)
Exploratory
• PK/PD (cema-cel and ALLO-647)
• Immunogenicity
• PROs

[a] Assessed per Lugano 2014 criteria. [b] EFS per IRC, PFS per IRC, and OS will undergo hierarchical testing.
Cema-cel, cemacabtagene ansegedleucel; EFS, event-free survival; IRC, independent review committee; MRD, minimal residual disease; OS, overall survival; PFS, progression-free survival; PK/PD, pharmacokinetics/pharmacodynamics; PRO, patient-reported outcome.

Figure 3: ALPHA3 Study Design

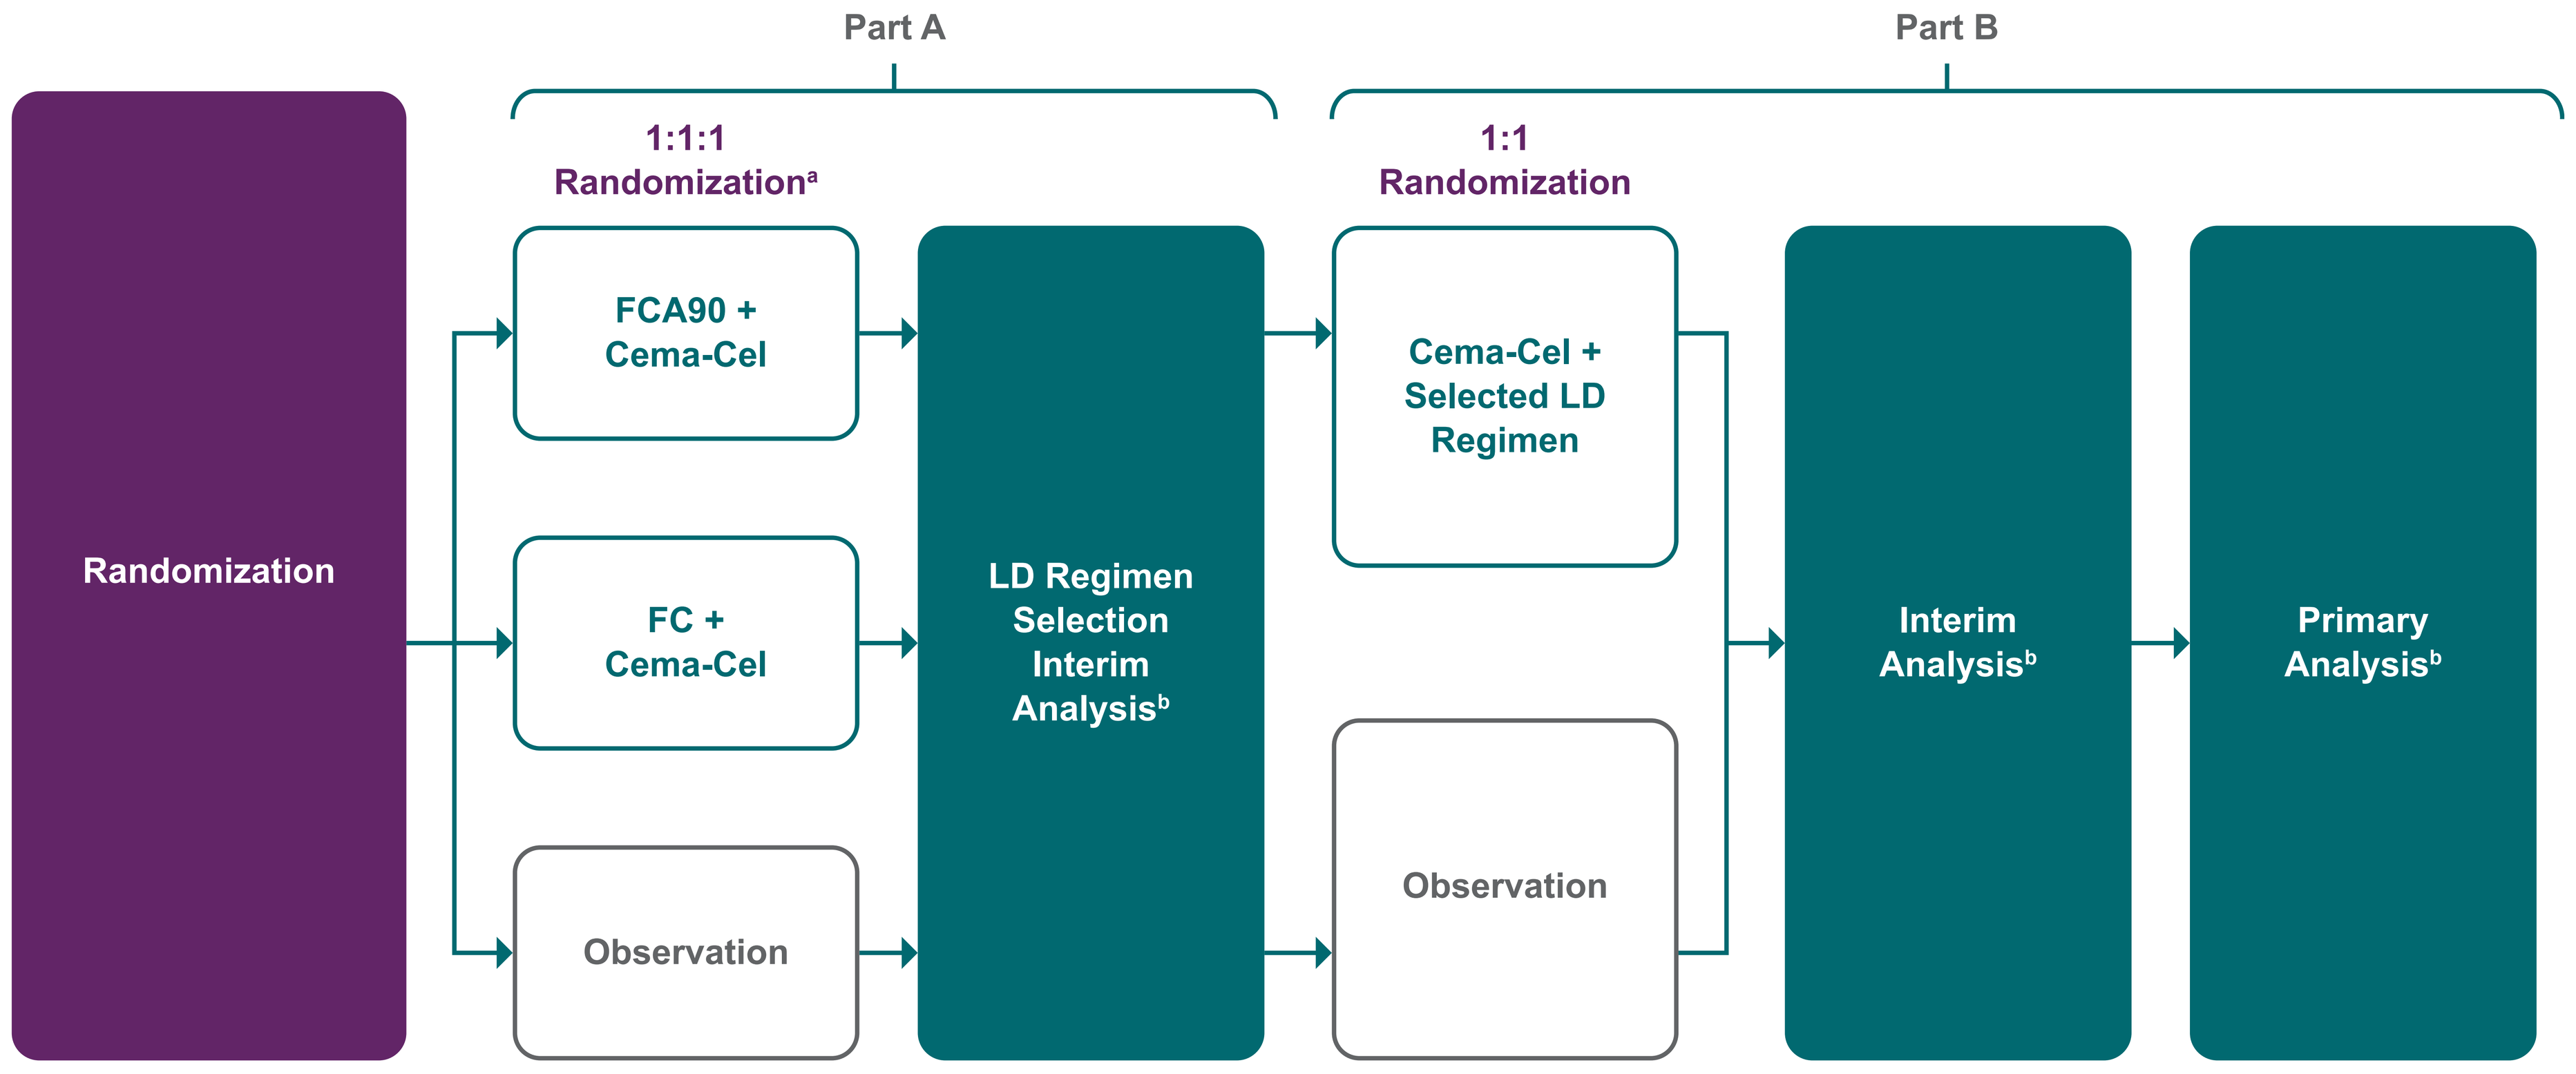
Part A
Part B
Randomization
1:1:1 Randomization[a]
FCA90 + Cema-Cel
FC + Cema-Cel
Observation
LD Regimen Selection Interim Analysis[b]
1:1 Randomization
Cema-Cel + Selected LD Regimen
Observation
Interim Analysis[b]
Primary Analysis[b]

[a] Randomization ratio may be adjusted after the safety interim analysis. [b] Safety and interim efficacy analyses will occur and culminate in LD regimen selection. Patients treated with the selected regimen or followed in observation during Part A will be included in inferential testing in Part B.
Cema-cel, cemacabtagene ansegedleucel; FC, fludarabine and cyclophosphamide; FCA90, fludarabine and cyclophosphamide and ALLO-647 (90 mg); LD, lymphodepletion.

Figure 4: ALPHA3 Study Patient Journey

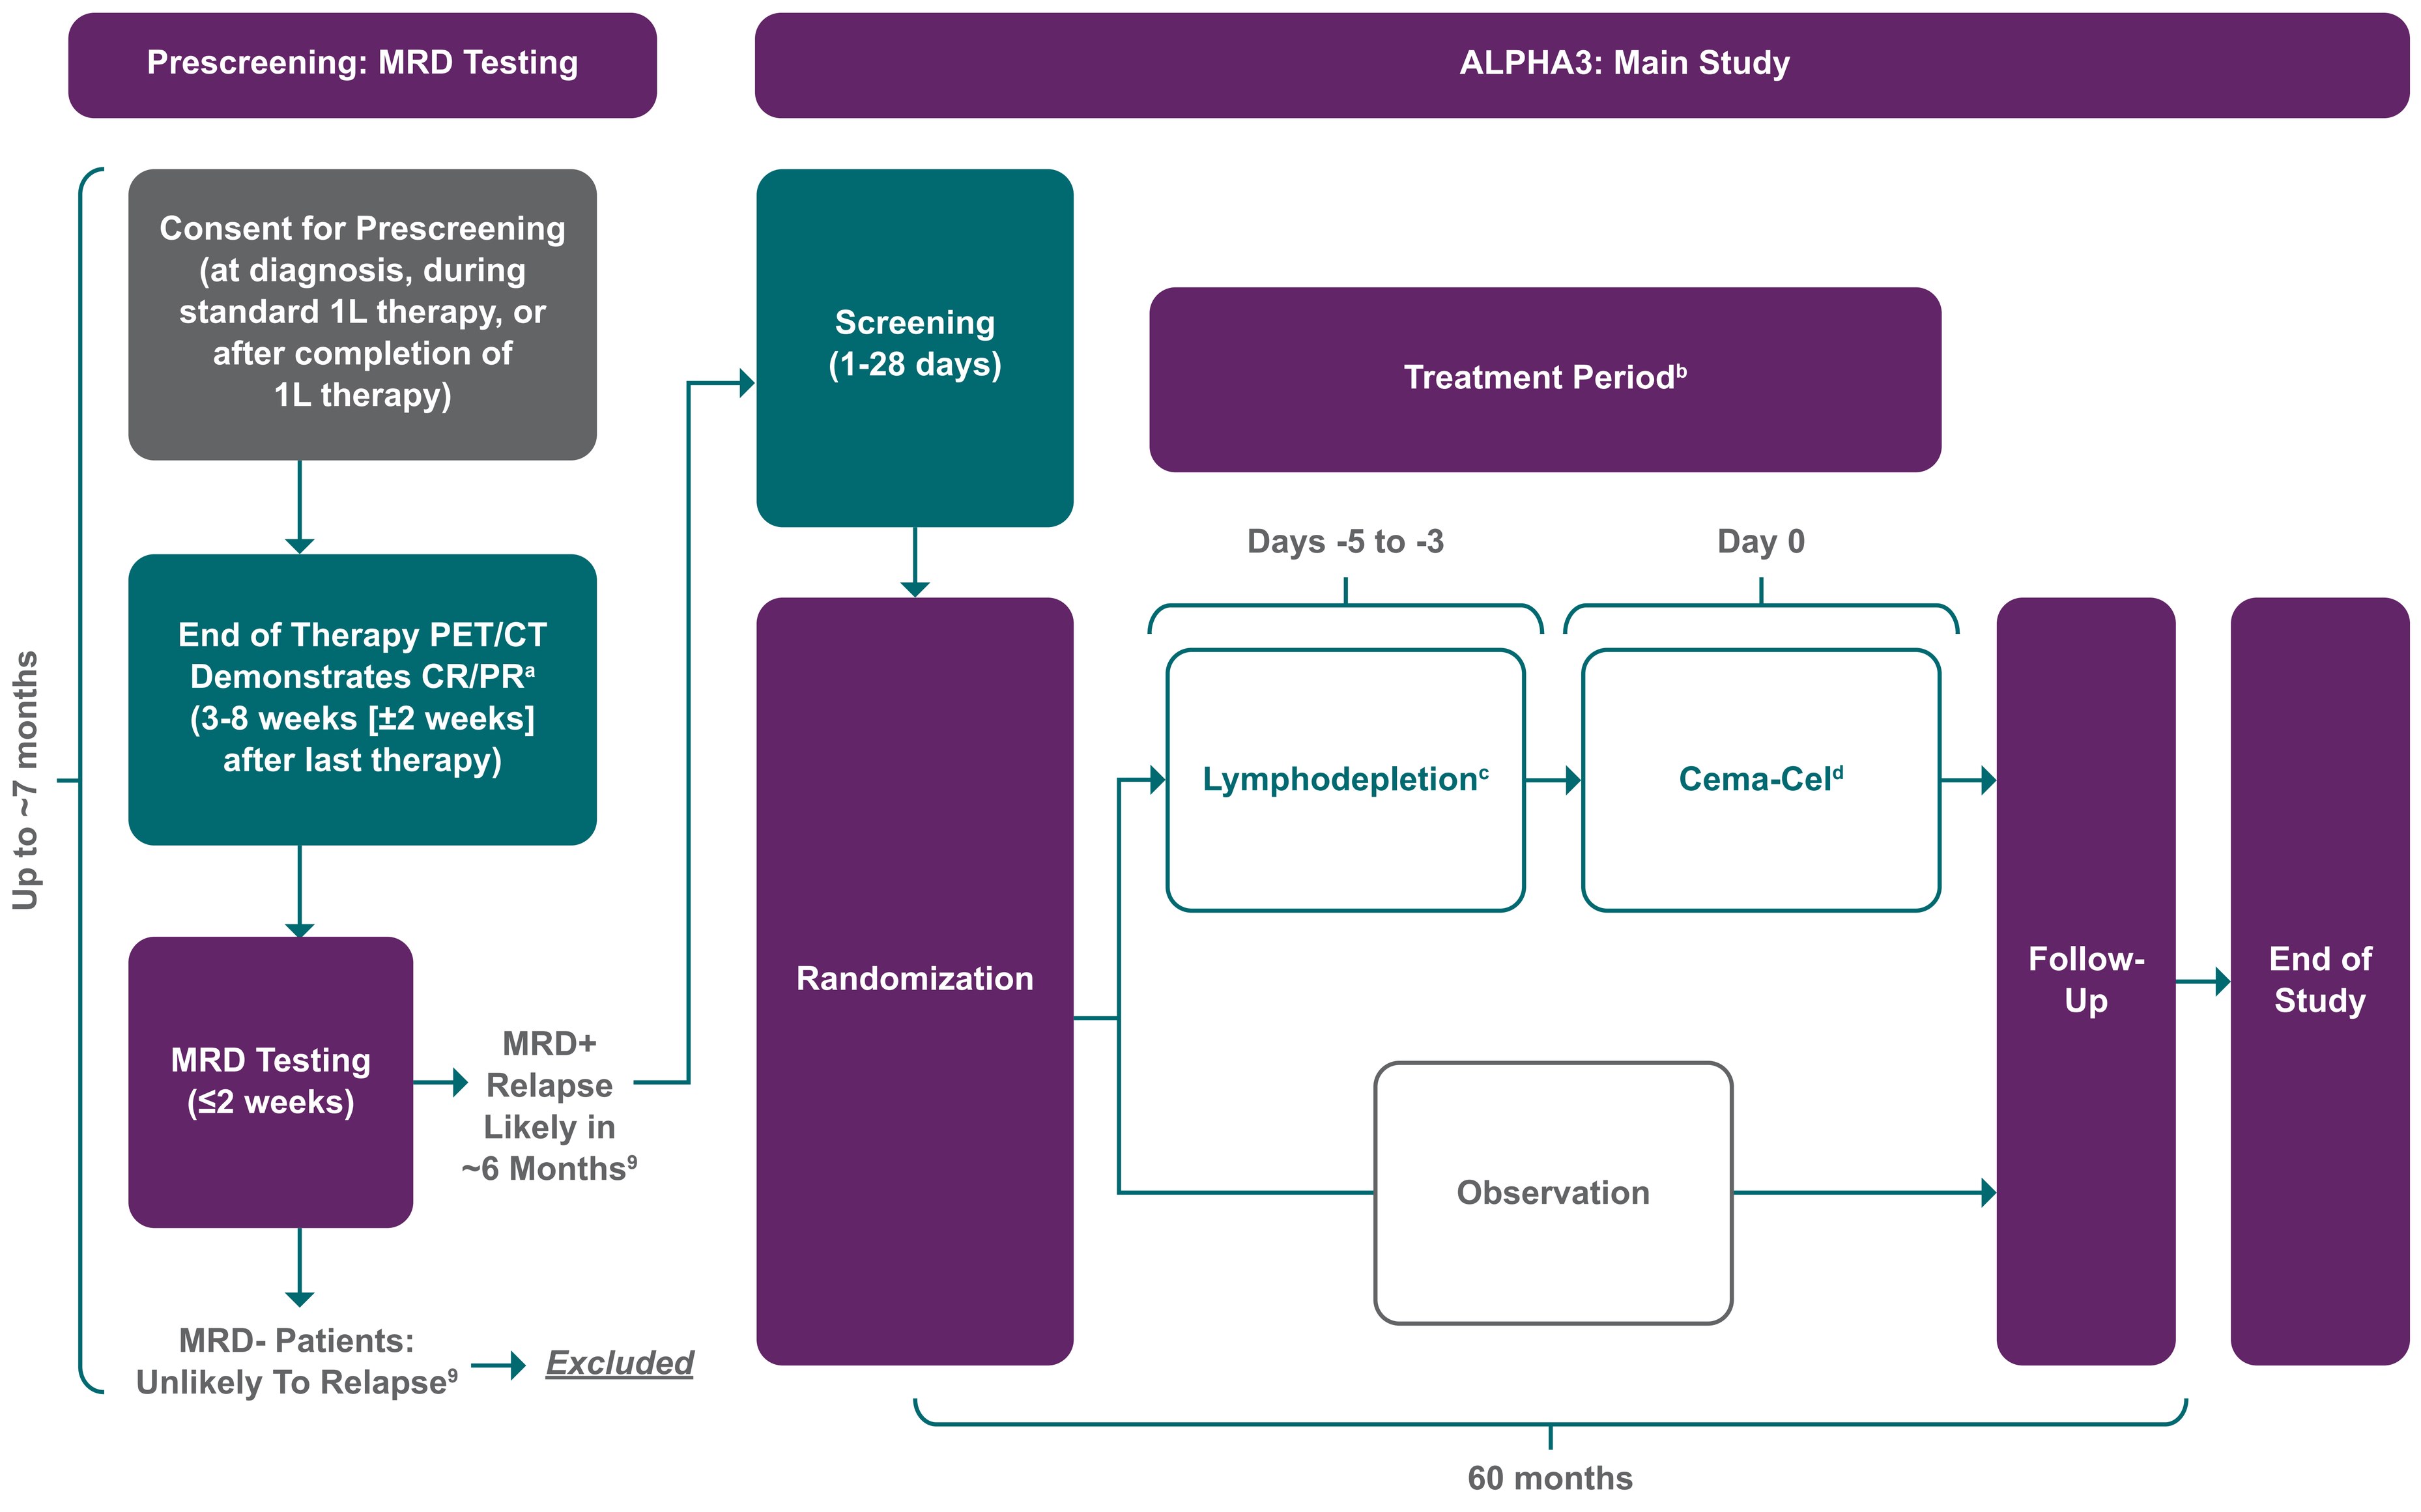
Prescreening: MRD Testing
ALPHA3: Main Study
Up to ~7 months
Consent for Prescreening (at diagnosis, during standard 1L therapy, or after completion of 1L therapy)
End of Therapy PET/CT Demonstrates CR/PR[a] (3-8 weeks [±2 weeks] after last therapy)
MRD Testing (≤2 weeks)
MRD+ Relapse Likely in ~6 Months[9]
MRD- Patients: Unlikely To Relapse[9]
Excluded
Screening (1-28 days)
Randomization
Treatment Period[b]
Days -5 to -3
Day 0
Lymphodepletion[c]
Cema-Cel[d]
Observation
Follow-Up
End of Study
60 months

[a] PR suitable for observation is defined as follows: all remaining PET-avid lesions have a Deauville score ≤5 by investigator assessment on end-of-therapy PET and intent to observe the patient without further treatment (including radiation therapy) until definitive disease progression. [b] Treatment can be done in the inpatient or outpatient setting. [c] The 3-day lymphodepletion regimen will consist of fludarabine (30 mg/m^2/day), cyclophosphamide (300 mg/m^2/day), and may include ALLO-647 (30 mg/day). [d] 120×10^6 CAR T cells.
1L, first line; CAR, chimeric antigen receptor; cema-cel, cemacabtagene ansegedleucel; CR, complete response; CT, computed tomography; MRD, minimal residual disease; PET, positron emission tomography; PR, partial response.

Table 2: Key Study Eligibility Criteria

Criteria for MRD Testing	Key Inclusion Criteria	Key Exclusion Criteria
Histologically confirmed diagnosis of DLBCL,[a] HGBCL, or PMBCL and at least one of the following clinical criteria: IPI score of 2-5, Ann Arbor Stage III or IV disease, equivocal response at interim or EOT PET/CT	MRD+ by ctDNA-based testing	Prior treatment with CD19-targeted therapies
Completed a full course of standard 1L therapy (must have included an anthracycline and an anti-CD20 monoclonal antibody), for example: • R-CHOP • Pola-R-CHP • DA-EPOCH-R	ECOG PS of 0 or 1	History of CNS involvement, transformation from other malignancy (transformed FL or MZL, or Richter's transformation), or T-cell/histiocyte-rich LBCL
Achieved a CR or PR suitable for observation[b] at the completion of 1L treatment based on PET/CT evaluation per Lugano 2014 criteria	No progression since MRD testing; response on PET/CT remaining within 5 weeks of randomization	History of clinically significant CNS dysfunction (eg, seizure disorder [uncontrolled in last 12 months], cerebrovascular ischemia/hemorrhage, dementia, cerebellar disease, cerebral edema)
MRD samples available for submission • Adequate tumor specimen from initial diagnosis • Adequate blood sample collected after final 1L treatment	Adequate hematologic, cardiac, pulmonary, renal, and hepatic function	Ongoing treatment with systemic immunosuppressive agents within 2 weeks or 5 half-lives (whichever is shorter) prior to enrollment[c]

[a] Includes DLBCL not otherwise specified, Epstein-Barr virus+ DLBCL, DLBCL with *IRF4/MUM1* rearrangement; high-grade B-cell lymphoma; and primary mediastinal B-cell lymphoma. [b] PR suitable for observation is defined as follows: all remaining PET-avid lesions have a Deauville score ≤5 by investigator assessment on end-of-therapy PET and intent to observe the patient without further treatment (including radiation therapy) until definitive disease progression. [c] Corticosteroids for physiologic replacement (<10 mg/day of prednisone equivalents) is acceptable.
1L, first line; CNS, central nervous system; CR, complete response; CT, computed tomography; DA-EPOCH-R, dose-adjusted rituximab, etoposide, prednisone, vincristine, cyclophosphamide, and doxorubicin; DLBCL, diffuse large B-cell lymphoma; ECOG PS, Eastern Cooperative Oncology Group performance status; EOT, end of therapy; FL, follicular lymphoma; HGBCL, high-grade B-cell lymphoma; IPI, International Prognostic Index; LBCL, large B-cell lymphoma; MRD, minimal residual disease; MZL, marginal zone lymphoma; PET, positron emission tomography; PMBCL, primary mediastinal B-cell lymphoma; Pola-R-CHP, polatuzumab vedotin, rituximab, cyclophosphamide, doxorubicin, and prednisone; PR, partial response; R-CHOP, rituximab, cyclophosphamide, doxorubicin, vincristine, and prednisone.

STUDY STATUS AND CONTACTS

- Approximately 240 patients will be enrolled at both academic- and community-based cancer centers
- Site activation in the US is nearing completion with international sites beginning to open this summer
- For more information, please visit www.clinicaltrials.gov
- Contact email: ALPHA3@allogene.com

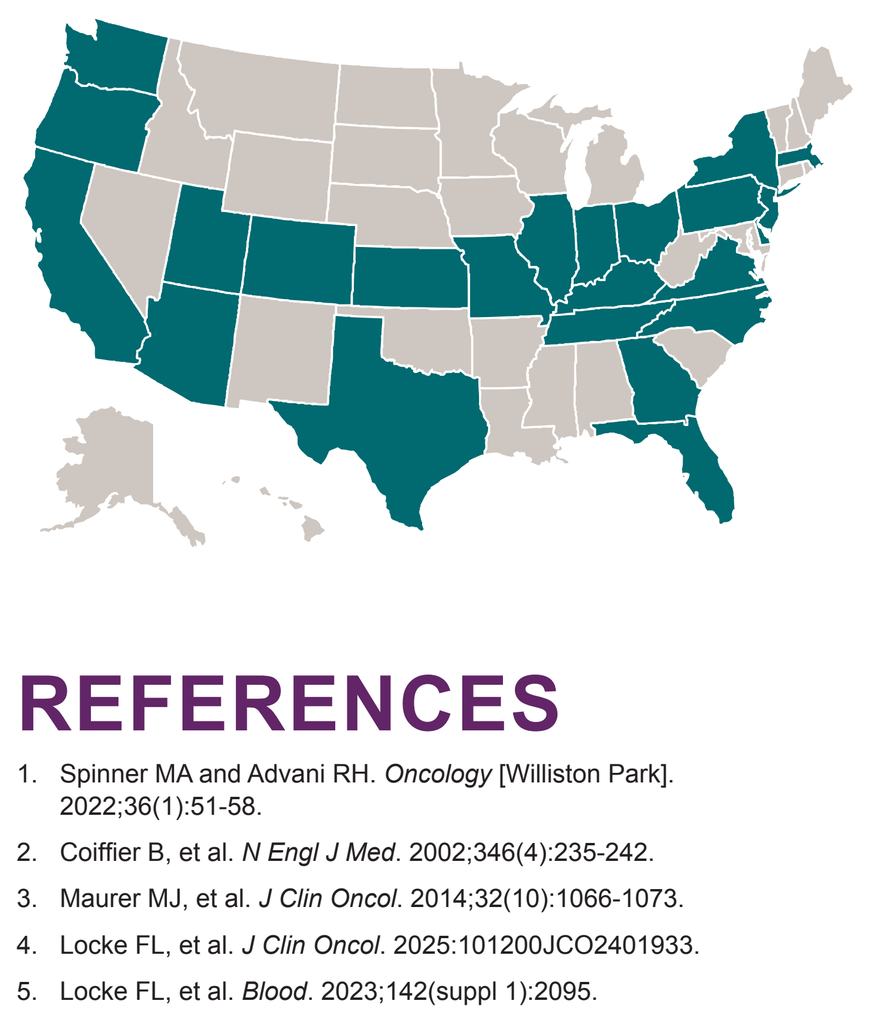

Anticipated Study Locations

Arizona	Illinois	New Jersey	Tennessee
California	Indiana	New York	Texas
Colorado	Kansas	North Carolina	Utah
Delaware	Kentucky	Ohio	Virginia
Florida	Massachusetts	Oregon	Washington
Georgia	Missouri	Pennsylvania	Washington, DC

REFERENCES

1. Spinner MA and Advani RH. *Oncology* [Williston Park]. 2022;36(1):51-58.
2. Coiffier B, et al. *N Engl J Med.* 2002;346(4):235-242.
3. Maurer MJ, et al. *J Clin Oncol.* 2014;32(10):1066-1073.
4. Locke FL, et al. *J Clin Oncol.* 2025:101200JCO2401933.
5. Locke FL, et al. *Blood.* 2023;142(suppl 1):2095.
6. Wudhikarn K, et al. *Blood Adv.* 2023;7(13):3192-3198.
7. Locke FL, et al. *Blood Adv.* 2020;4(19):4898-4911.
8. Perica K, et al. *Biol Blood Marrow Transplant.* 2018;24(6):1135-1141.
9. Roschewski M, et al. ICML 2023. Oral 112.

ACKNOWLEDGMENTS

Allogene's investigational AlloCAR T™ oncology products utilize Cellectis technologies. These products are developed based on an exclusive license granted by Cellectis to Servier. Servier, which has an exclusive license to the anti-CD19 AlloCAR T™ investigational products from Cellectis, has granted Allogene exclusive rights to these products in the U.S., all EU Member States and the United Kingdom.

Medical writing and editorial assistance were provided by Second City Science and funded by Allogene Therapeutics, Inc.

©American Society of Hematology (2024). Reused with permission.

Contact Allogene Therapeutics for permission to reprint and/or distribute.

CONTACT INFORMATION

Jason Westin: jwestin@mdanderson.org

Presented at the American Society of Clinical Oncology Annual Meeting, May 30-June 3, 2025; Chicago, IL & Online